Nighttime Symptom Experience and Treatment Challenges: Perspectives From People With Narcolepsy

Luis E. Ortiz, MD[1]; Maggie Lavender, MSN, RN, FNP-C[2,3]; Anne Marie Morse, DO[4]; Beth Schneider, BA[5]; Matthew Horsnell, BS[6]; Dan Liss[7]; Shawn Kuczenski[8]; Jennifer Gudeman, PharmD[9,a]

[1]Johns Hopkins Medical Institutions, Johns Hopkins All Children's Hospital, St. Petersburg, FL, USA; [2]Comprehensive Sleep Medicine Associates, Houston, TX, USA; [3]Sleep Medicine Advanced Practice Providers (SMAPP) President and Founder, East Hanover, NJ, USA; [4]Geisinger Commonwealth School of Medicine, Geisinger Medical Center, Janet Weis Children's Hospital, Danville, PA, USA; [5]Swoop, New York, NY, USA; [6]Patient author, Erin, TN, USA; [7]Patient author, New Oxford, PA, USA; [8]Patient author, Perryville, AR, USA; [9]Avadel Pharmaceuticals, Chesterfield, MO, USA; [a]At the time of the study

Conclusions

- Nighttime symptoms, including disturbed nocturnal sleep, insomnia, and REM-related phenomena, are common narcolepsy features and have a negative impact on the daily lives of PWN
 - PWN seek relief from both daytime and nighttime symptoms of narcolepsy
- Although the experiences and needs of PWN extend beyond daytime sleepiness, more than one-third of respondents had not discussed nighttime treatment options with their clinician that could improve nighttime sleep
 - Greater understanding of both daytime and nighttime symptoms experienced by PWN may help clinicians better support, educate, and treat their patients

Introduction

- Narcolepsy is a rare, chronic sleep disorder characterized by daytime and nighttime symptoms, including excessive daytime sleepiness (EDS), disturbed or disrupted nocturnal sleep, and rapid eye movement (REM)-related phenomena such as sleep-related hallucinations, sleep paralysis, and cataplexy in persons with narcolepsy type 1 (NT1)[1,2]
- Symptoms of narcolepsy can be burdensome and negatively affect quality of life for people with narcolepsy (PWN)[3]
- Sodium oxybate (SXB) is a strong recommendation by the American Academy of Sleep Medicine for the treatment of EDS, cataplexy, and disease severity associated with narcolepsy[4]
 - Immediate-release SXB and calcium/magnesium/potassium/sodium (mixed-salt) oxybates require twice-nightly dosing, with the second dose administered 2.5-4 hours after the first dose[5,6]
 - Extended-release, once-nightly SXB (ON-SXB) is administered once at bedtime[7]
- As narcolepsy is fundamentally a 24-hour disorder, it is important to assess the perspectives of PWN regarding their experience with both daytime and nighttime symptoms and their treatment options to ensure adequate management of all symptoms

Objective

- To identify and describe common symptoms and daytime and nighttime challenges of PWN and gain insights among those experienced with ON-SXB and oxybate-naive PWN

Methods

- MyNarcolepsyTeam is an online social network where >12,000 members can share their experiences, challenges, journeys, and questions related to narcolepsy in their own words[3,8]
- This study used a 2-pronged approach in which MyNarcolepsyTeam members from the US participated in 1) an online survey and/or 2) in-depth online focus group discussions
- Participants were required to indicate whether they had NT1, had narcolepsy type 2 (NT2), or were unsure of their narcolepsy type

ONLINE SURVEY

- In September 2022, PWN were invited by email to complete an anonymous, 47-question online survey on topics including questions on symptoms, treatment regimens, challenges, and desired treatment outcomes
 - The survey included a variety of question/response formats, including Yes/No, ranking, and multiple-choice selection (with selection of ≥1 option allowed)
 - For the "most troubling symptoms" question, respondents selected ≥1 option from a 19-symptom list of patient-friendly terms (definitions were not provided)
- This study qualified for institutional review board (IRB) exemption

ONLINE FOCUS GROUP DISCUSSION

- Separately, PWN were invited to participate in 1 of 2 small online focus group discussion panels
 - 1 discussion including ON-SXB–experienced PWN was held in July 2025
 - 1 discussion including oxybate-naive PWN was held in September 202
- Both discussion panels were led by a facilitator and conducted using a structured guide informed by findings from previous studies and themes that emerged from earlier discussions
 - Illustrative patient quotes were selected based on relevance to the discussion topic

Results

ONLINE SURVEY

- Of 109 respondents, the majority were female (78%; n=85) and aged ≥50 years (62%; n=68); 41% (n=45) had NT1, 40% had narcolepsy type 2 (NT2; n=44), and narcolepsy type was unknown for 18% (n=20) of respondents
- 97% (n=106) of respondents indicated narcolepsy had a "great deal" or "a lot" of impact on their daily life
 - 93% (42/45) of individuals with NT1 and 100% (44/44) of those with NT2 indicated narcolepsy had "a great deal" or "a lot" of impact
- Symptoms reported as most troubling included EDS (90%), fatigue (84%), nighttime sleep disturbances (81%), and cognitive/memory problems (80%)
 - Nighttime sleep disturbances included poor-quality sleep (67%), disrupted/fragmented sleep (62%), insomnia (40%), or frequent awakenings (40%)
 - Cataplexy was reported as the most troubling symptom among 64% (29/45) of respondents with NT1
- The most important reported potential treatment benefits were stopping EDS (99%), allowing productivity (96%), improving well-being (95%), increasing energy level (95%), and improving memory/cognition (90%; **Figure 1**)
- Most respondents desired treatments that improved sleep quality (85%) and helped them sleep through the night (70%); over a third of respondents desired treatments that stopped REM-related phenomena (36%-39%) **(Figure 1)**
 - However, only 64% had discussed nighttime treatment options with their clinician **(Figure 2)**
- Recommendations for nighttime treatment options were more common for respondents seeing a sleep specialist (54%) compared with those seeing a generalist (41%)
- Oxybates were the most frequent clinician-recommended nighttime treatment (69%), followed by melatonin (12%), zolpidem (8%), and sertraline (4%)
- Among respondents currently taking oxybates, their decision to start taking oxybates was most frequently influenced by hearing from their clinician about the effectiveness of oxybates (60% [12/20]) and that oxybates are the best medications for treating both daytime and nighttime symptoms (55% [11/20]; **Figure 3**)

EDS, excessive daytime sleepiness; SXB, sodium oxybate.

- Respondents taking oxybates were twice as likely to be satisfied or extremely satisfied with their current treatment regimen vs those not taking oxybates (70% [14/20] vs 37% [28/76]; **Figure 4**)

ONLINE FOCUS GROUP DISCUSSIONS

- The small, separately conducted focus group discussions (ON-SXB–experienced, n=3; oxybate-naive, n=5) qualitatively illustrate the survey results and the nighttime symptom burden experienced by PWN (ie, vivid dreams and fragmented sleep; **Figure 5**)
- Oxybate-naive PWN described challenges with daytime alertness following poor nighttime sleep and desired a treatment that would allow them to wake refreshed **(Figure 6)**
- ON-SXB–experienced PWN desired a treatment that would allow them to sleep through the night and wake refreshed **(Figure 7)**

Study Limitations

- These data were collected exclusively from survey responses; participants who are MyNarcolepsyTeam members and their responses may not be reflective of the broader population of PWN
- Additional limitations include the observational design, potential for recall bias, and small sample size
 - Small numbers of patients in focus group discussions may not be representative of the broader narcolepsy treatment population

FUNDING

This study was funded by Avadel Pharmaceuticals (Chesterfield, MO). Avadel Pharmaceuticals Limited (formerly Avadel Pharmaceuticals plc) is an affiliate of Alkermes plc.

ACKNOWLEDGMENTS

Medical writing support was provided by Jenna Kitz, PhD, from Citrus Health Group, Inc. (Chicago, IL) and was funded by Avadel Pharmaceuticals (Chesterfield, MO).

REFERENCES

1. Kornum BR, et al. Nat Rev Dis Primers. 2017;3(1):16100.
2. American Academy of Sleep Medicine. International Classification of Sleep Disorders. 3rd ed. American Academy of Sleep Medicine; 2014.
3. Ortiz LE, et al. CNS Drugs. 2025;39(suppl 1):23-36.
4. Maski K, et al. J Clin Sleep Med. 2021;17(9):1881-1893.
5. Xyrem® (sodium oxybate oral solution, CIII). Full Prescribing Information. Jazz Pharmaceuticals, Inc; 2025.
6. Xywav® (calcium, magnesium, potassium, and sodium oxybate). Full Prescribing Information. Jazz Pharmaceuticals, Inc; 2025.
7. LUMRYZ® (sodium oxybate for extended-release oral suspension, CIII). Full Prescribing Information. Avadel Pharmaceuticals; 2025.
8. MyNarcolepsyTeam. MyHealthTeam. Accessed December 08, 2025. https://www.mynarcolepsyteam.com

FIGURE 1. Desired Treatment Endpoints

Q. How important are each of the following potential benefits when considering a narcolepsy treatment? (n=109)

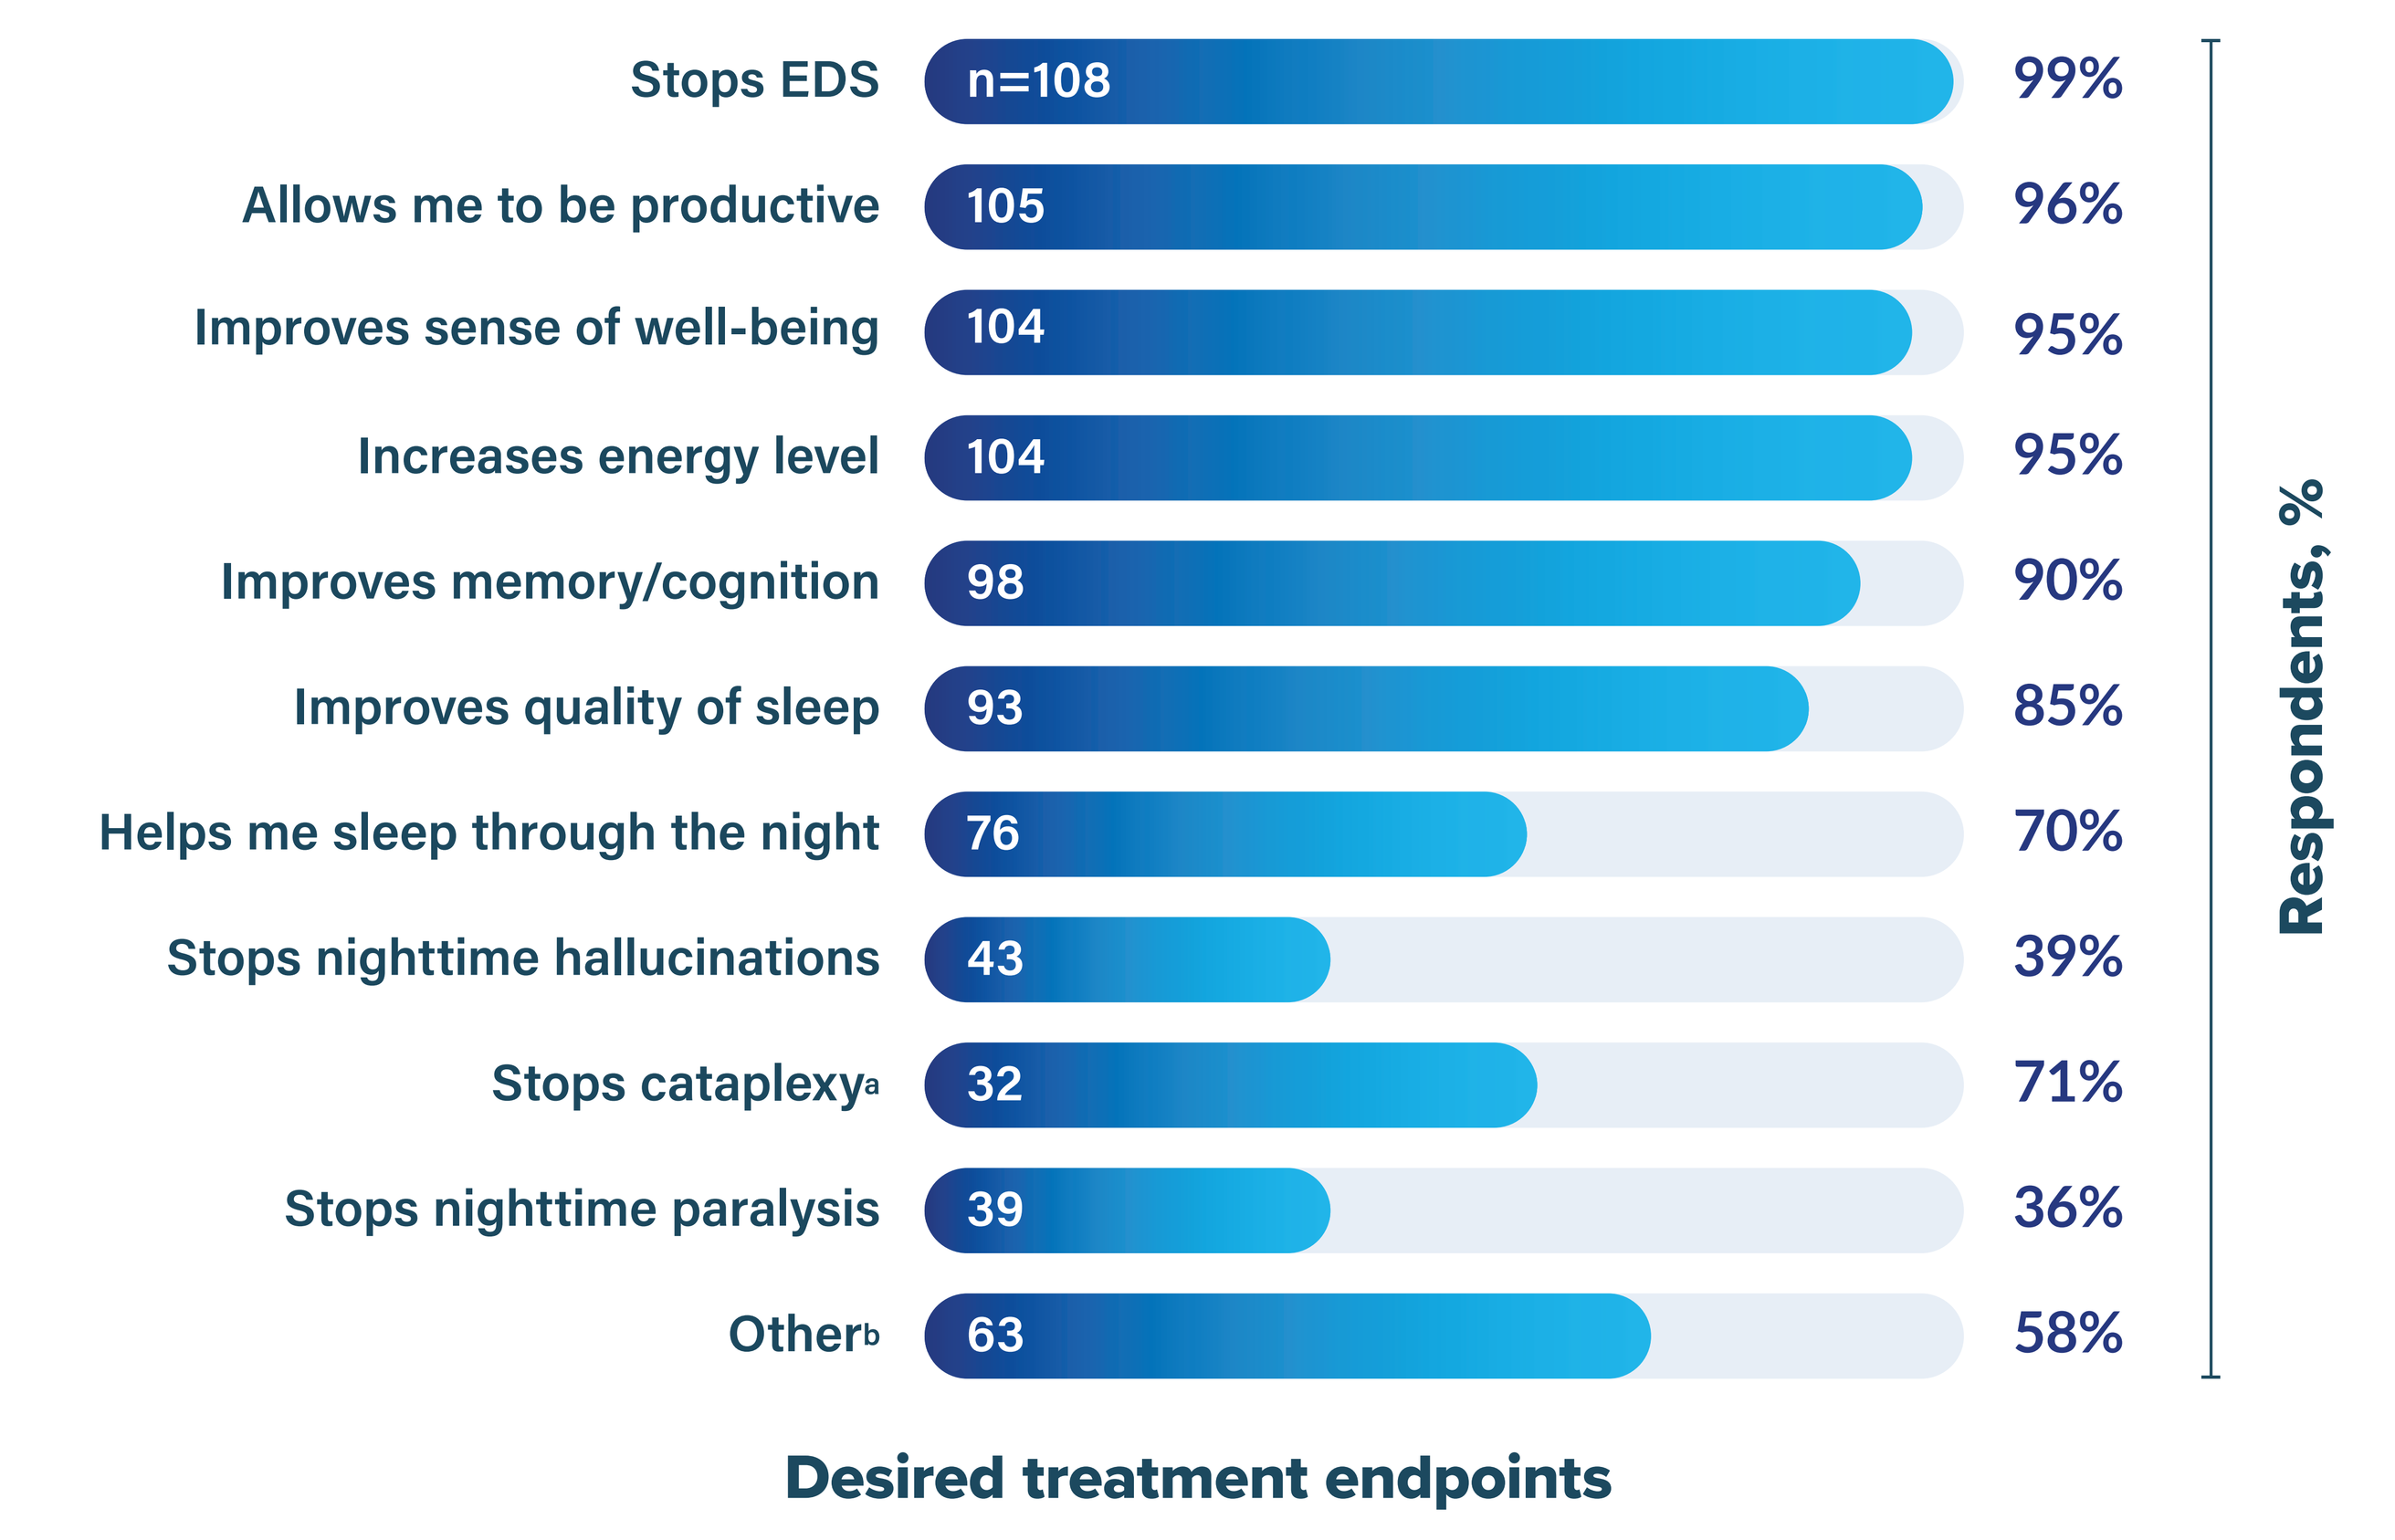
Stops EDS
n=108
99%
Allows me to be productive
105
96%
Improves sense of well-being
104
95%
Increases energy level
104
95%
Improves memory/cognition
98
90%
Improves quality of sleep
93
85%
Helps me sleep through the night
76
70%
Stops nighttime hallucinations
43
39%
Stops cataplexy[a]
32
71%
Stops nighttime paralysis
39
36%
Other[b]
63
58%
Respondents, %
Desired treatment endpoints

FIGURE 2. Experience Discussing Treatment Options With Clinicians

Q. Have you and your doctor ever discussed treatment options that could improve your ability to sleep at night? (Asked of those seeing a doctor; n=105)

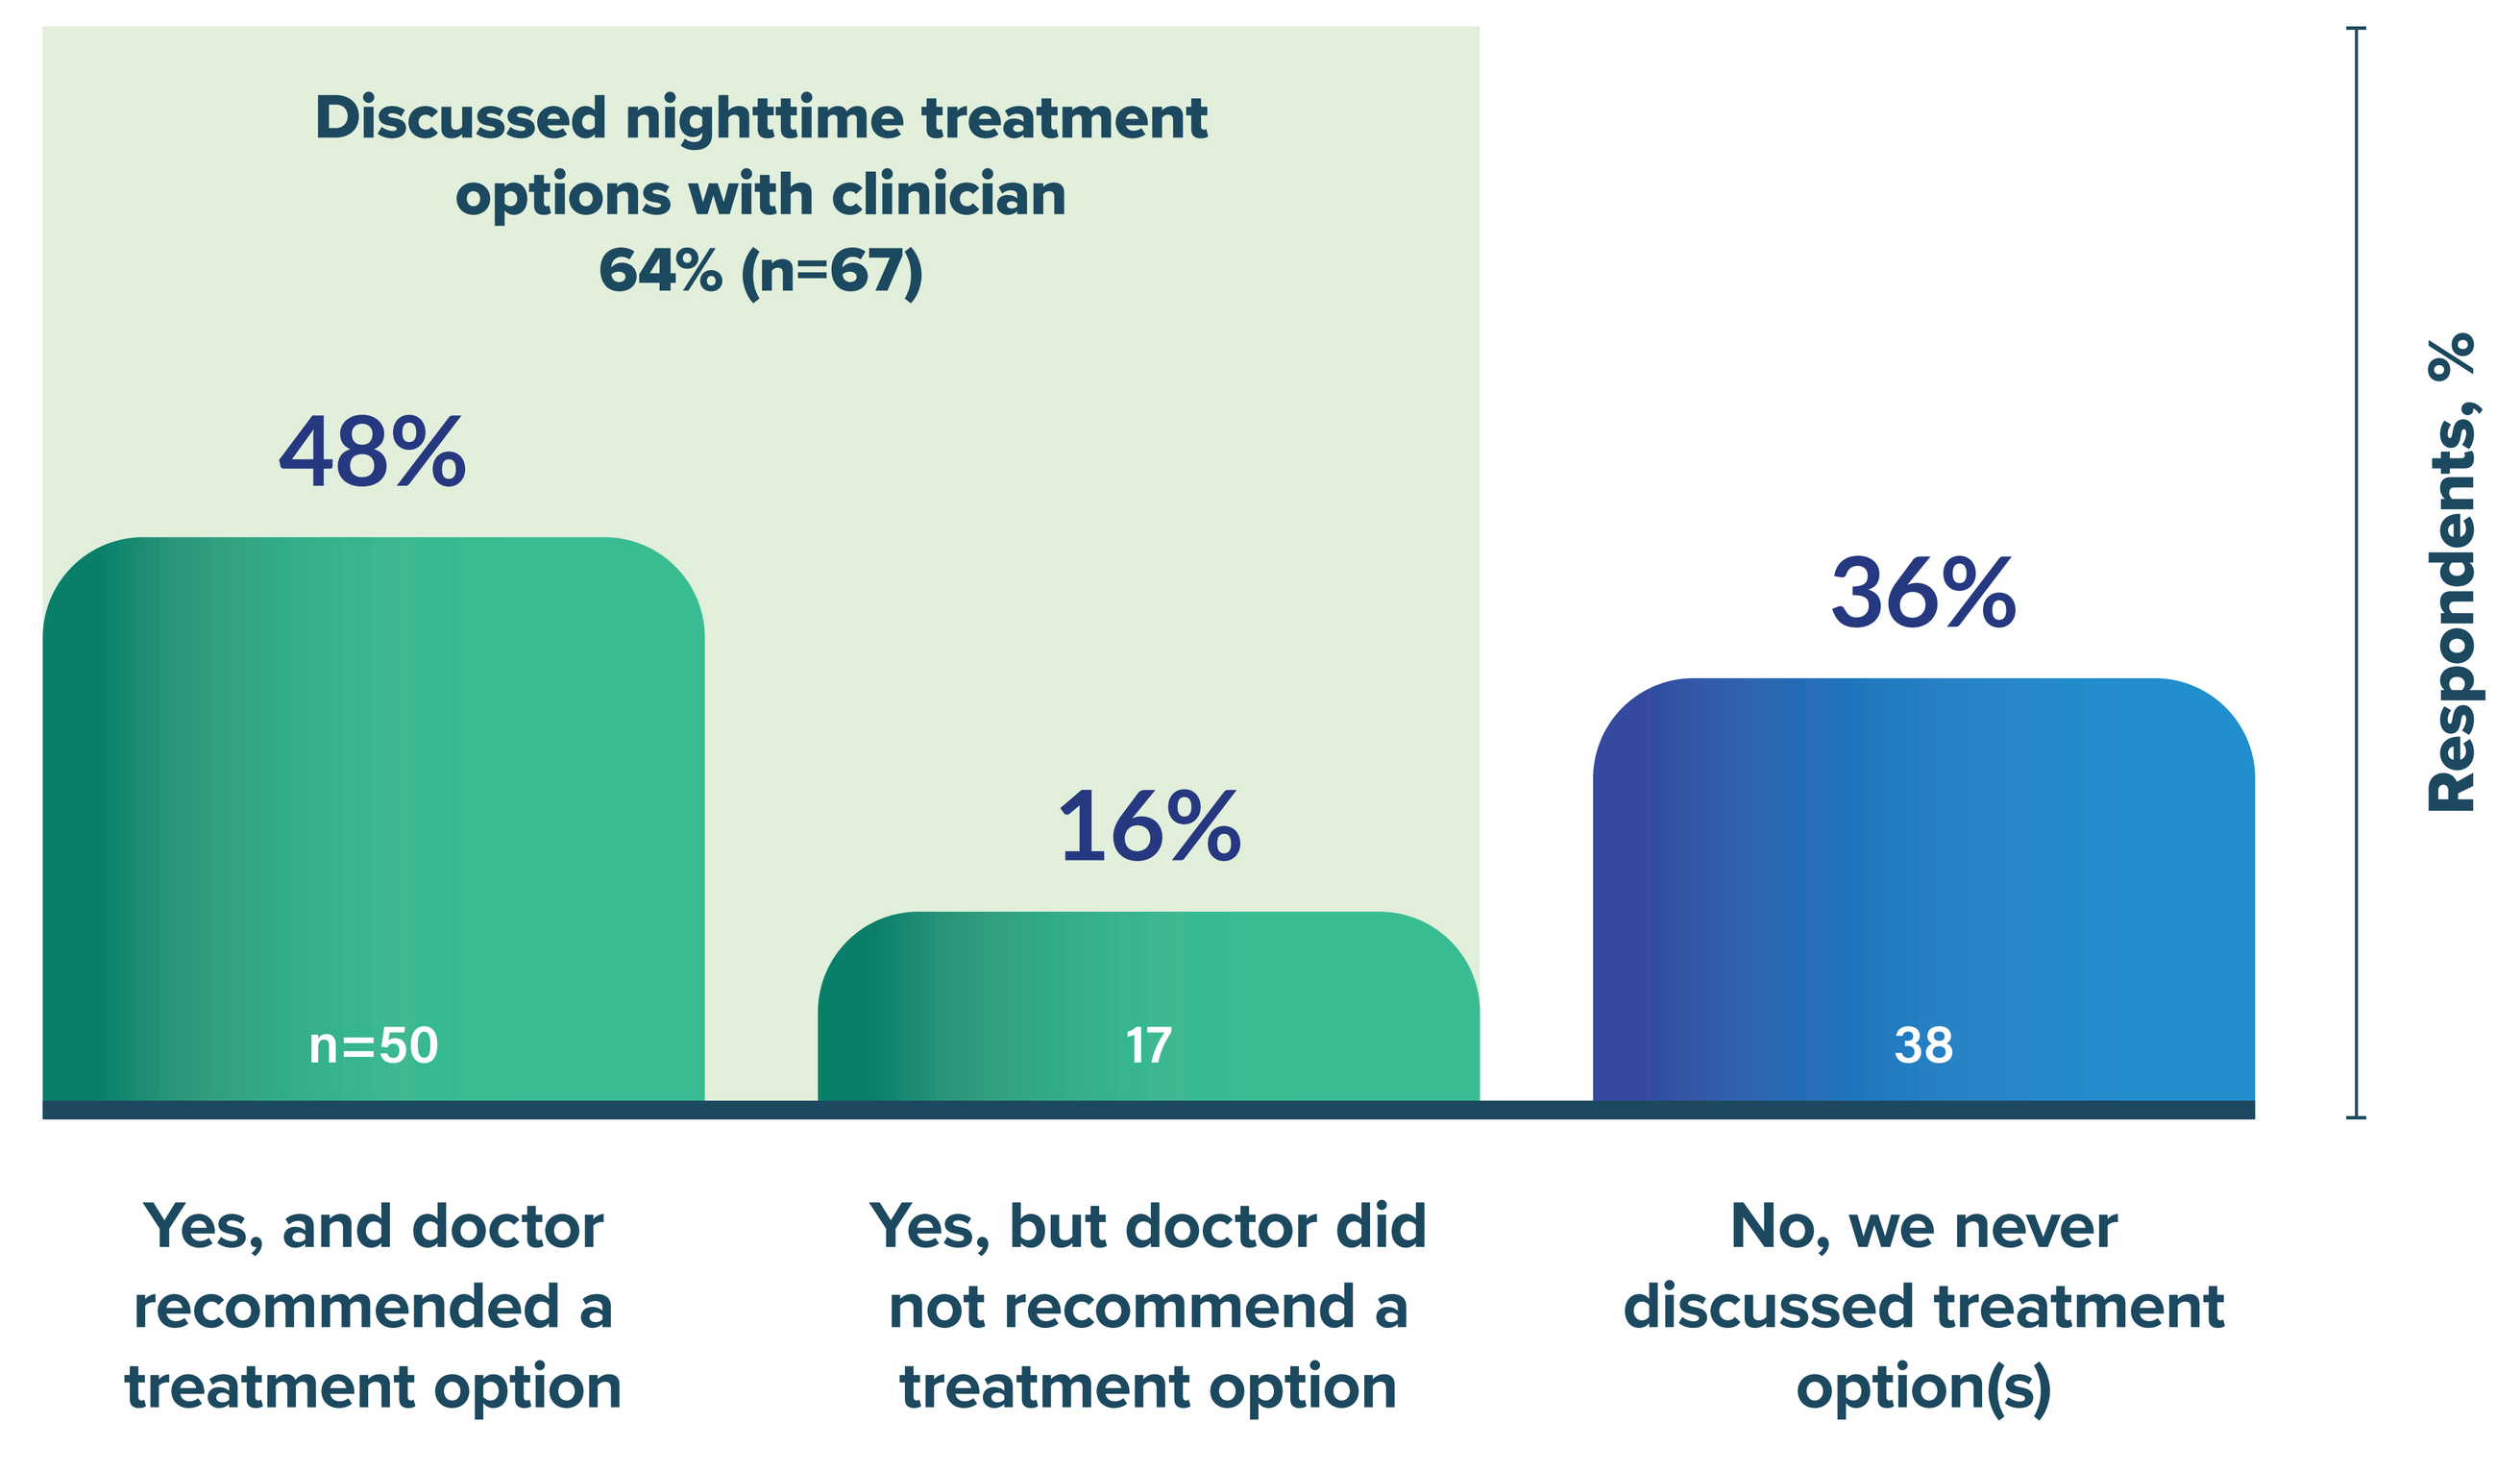
Discussed nighttime treatment options with clinician 64% (n=67)
48%
n=50
16%
17
36%
38
Respondents, %
Yes, and doctor recommended a treatment option
Yes, but doctor did not recommend a treatment option
No, we never discussed treatment option(s)

FIGURE 3. What Clinicians Said About SXB

Q. What did your doctor say to you that most helped you decide to get on sodium oxybate? Select all that apply (n=20)

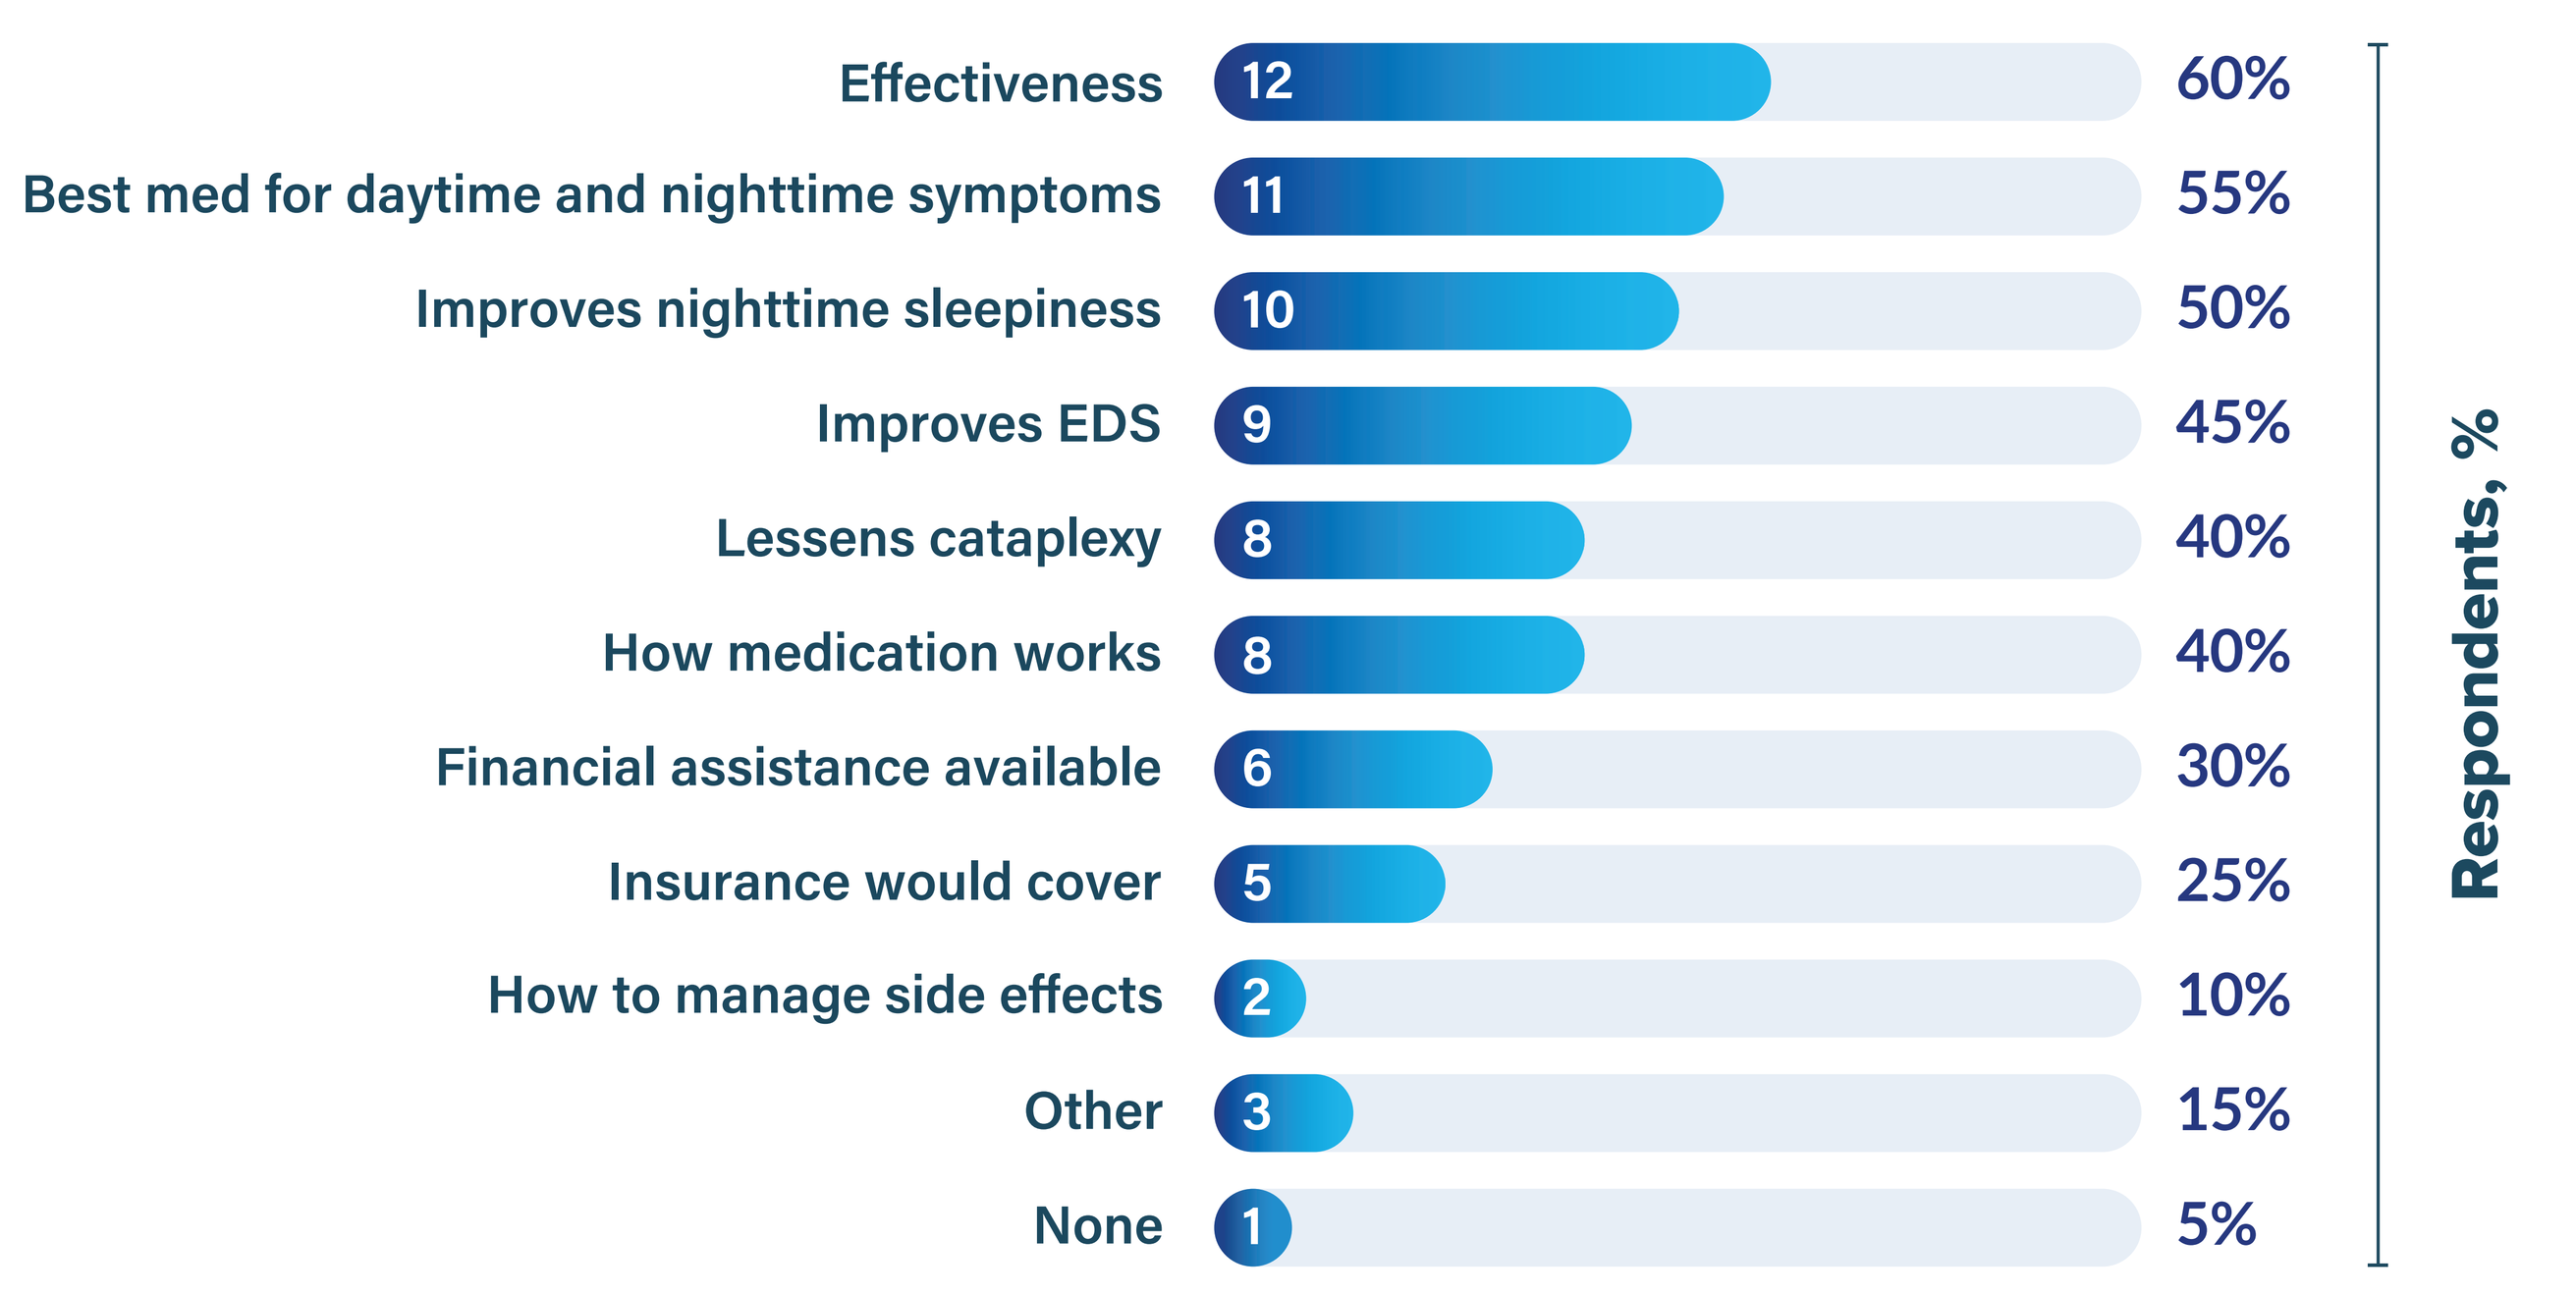
Effectiveness
12
60%
Best med for daytime and nighttime symptoms
11
55%
Improves nighttime sleepiness
10
50%
Improves EDS
9
45%
Lessens cataplexy
8
40%
How medication works
8
40%
Financial assistance available
6
30%
Insurance would cover
5
25%
How to manage side effects
2
10%
Other
3
15%
None
1
5%
Respondents, %

FIGURE 4. Satisfaction With Current Treatment Regimen

Q. Overall, how satisfied are you with your current medication regimen for treating narcolepsy?

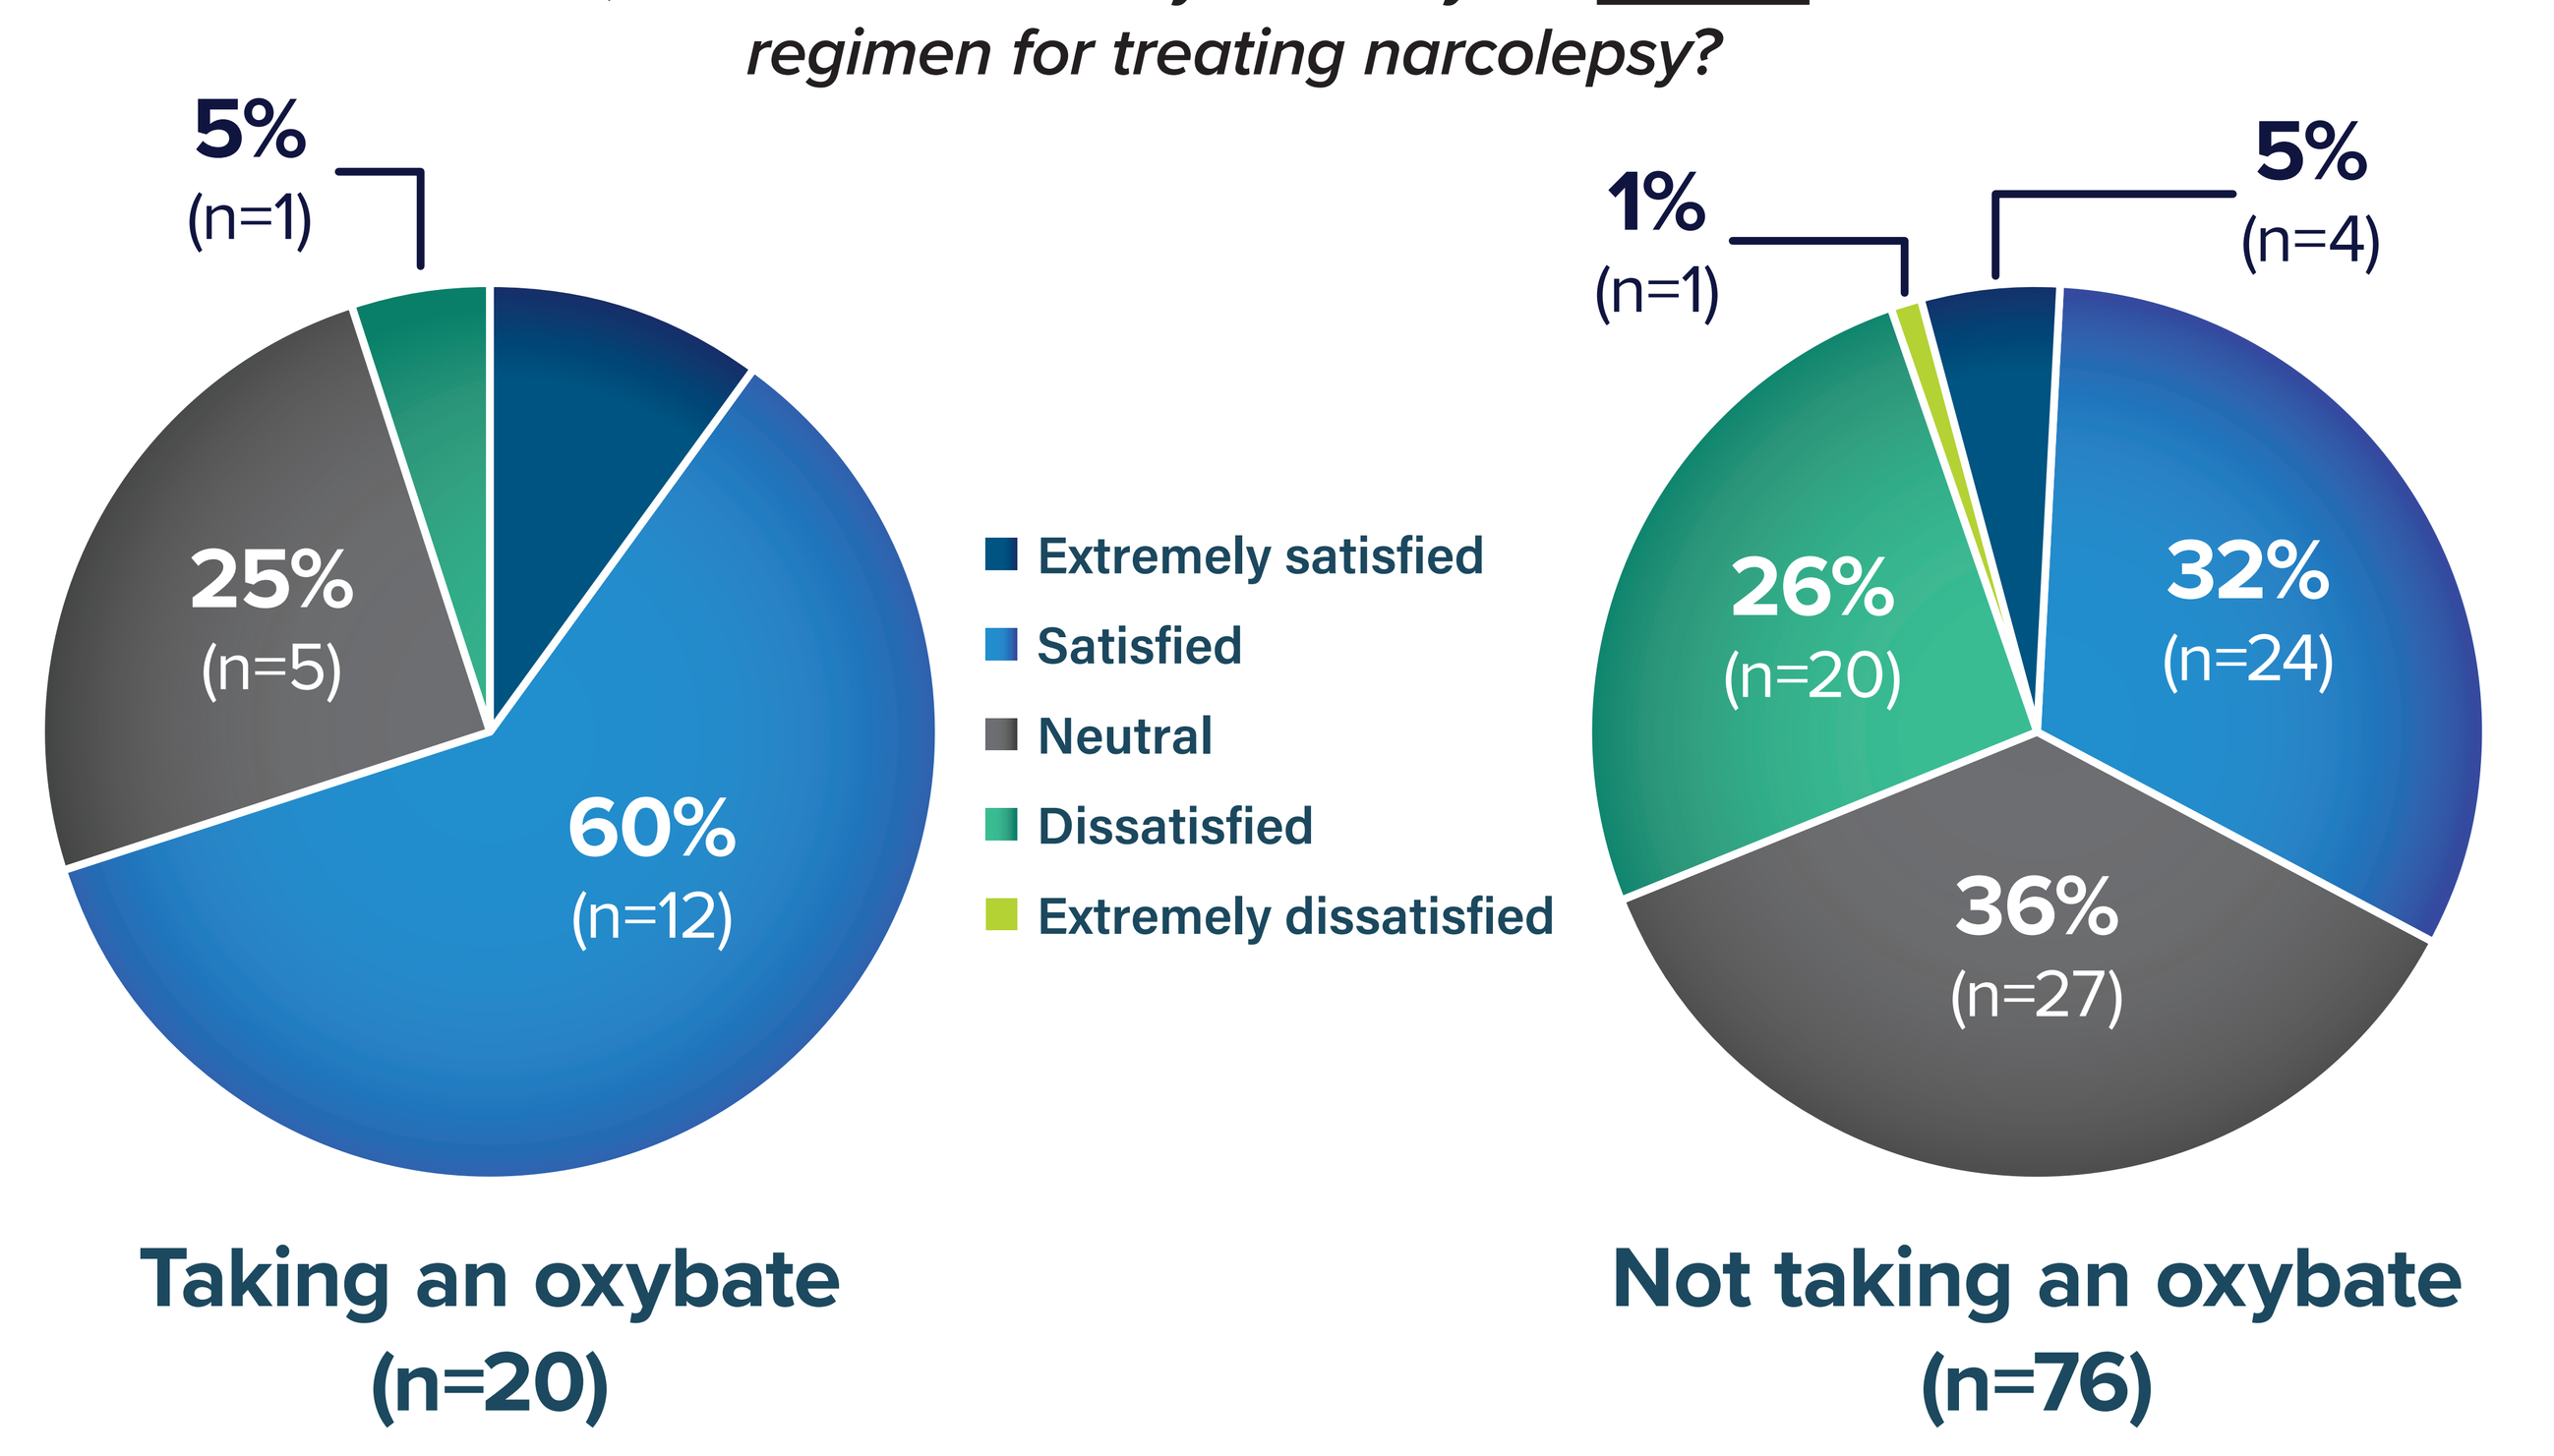
5%
(n=1)
25%
(n=5)
60%
(n=12)
Taking an oxybate (n=20)
Extremely satisfied
Satisfied
Neutral
Dissatisfied
Extremely dissatisfied
1%
(n=1)
5%
(n=4)
26%
(n=20)
32%
(n=24)
36%
(n=27)
Not taking an oxybate (n=76)

FIGURE 5. Patient Discussions on Nighttime Symptom Burden

Patient voices on vivid dreams

"The vivid dreaming can mess you up. Sometimes you have such vivid nightmares that sometimes you have to reassure yourself that it's not real. I wanted it to stop. Just to stop."

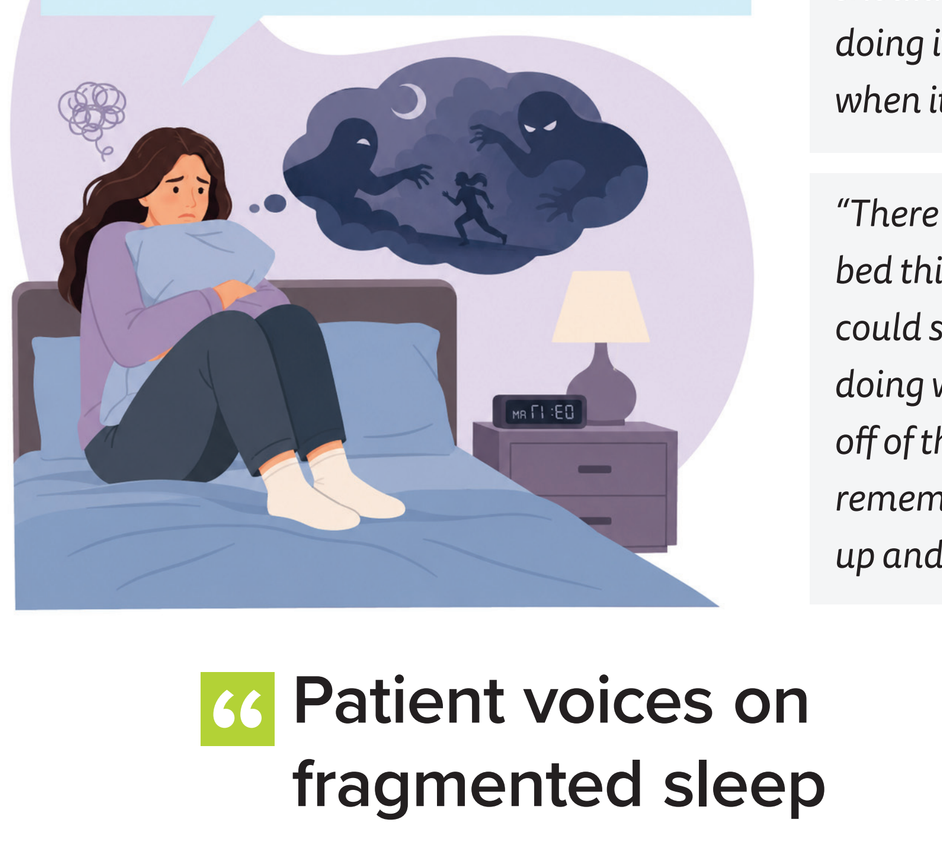

"I have these weird visions. I do talk in my sleep. I have vivid dreams. I've had one episode where you feel like you can't move, sleep paralysis."

"When the dreams first happened for me, it was very scary mentally. I was telling myself, you should get up. But physically, my body just wasn't doing it for me. And so it was a very scary thing when it first started. And again, you just adapt."

"There was night terrors standing on the foot of the bed thinking that my baby was in the ceiling and I could see her falling. I remember it felt like I was doing what I was supposed to do because I leaped off of the bed and gave myself a concussion. I remember the sleepwalking and then I would wake up and go, whoa, what's going on?"

Patient voices on fragmented sleep

"I would wake up after two minutes and people would say that I can't dream within that period, but my dreams were always lucid and I had some sleep paralysis. It felt distressing that there was a period of time I was so afraid to sleep. I would rely on other medicines to try and sleep. The longest time I would sleep would be about an hour, hour day. It was hard. It was hard. Some anxiety"

"I don't sleep that great. Maybe four hours at a stretch I can do, and then I have to get up and then I can fall back asleep."

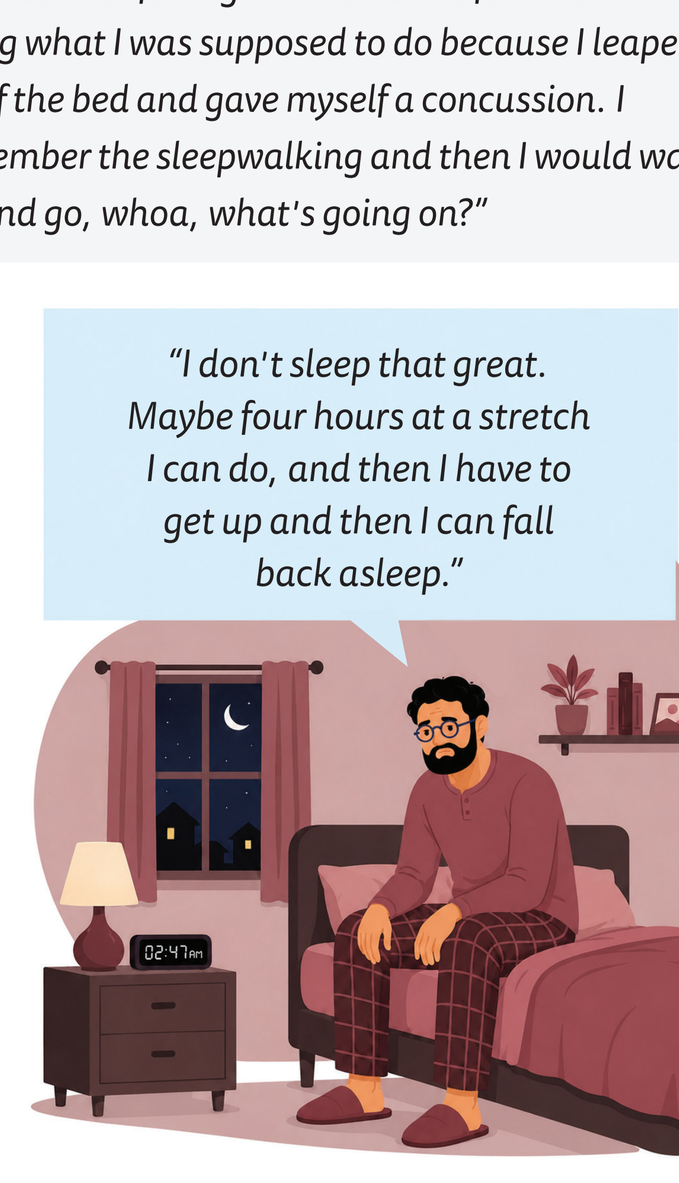

FIGURE 6. Challenges and Desired Treatment Outcomes in Oxybate-Naive PWN

Patient voices on challenges with daytime alertness

"There'll be some days if I don't have enough time on the pillow that I'll wake up and I'm just exhausted."

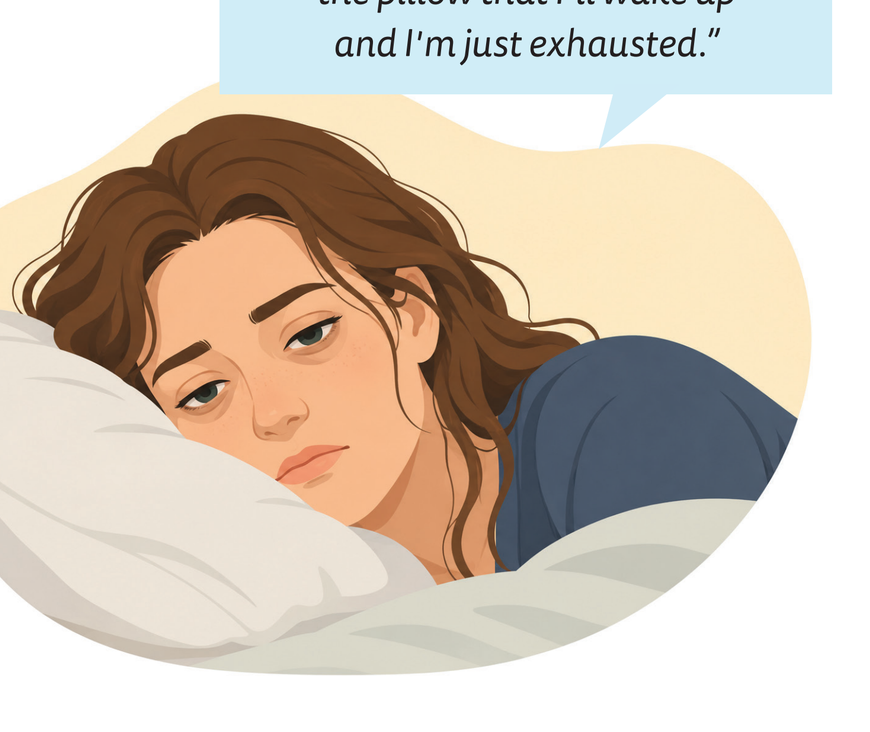

"I spend most of the day asleep or napping in and out. It's not even asleep. I get two to four hours of solid sleep maybe at night and that's it. I used to sleep 18 plus hours a day, and then one day it just switched and that was like two, four hours. And that's all I can sleep. I'm like every few minutes to every few hour, every hour. But at nighttime, I'm awake all night."

"Waking up refreshed? I wake and I'm about to eat breakfast and then I'm about to go back to sleep."

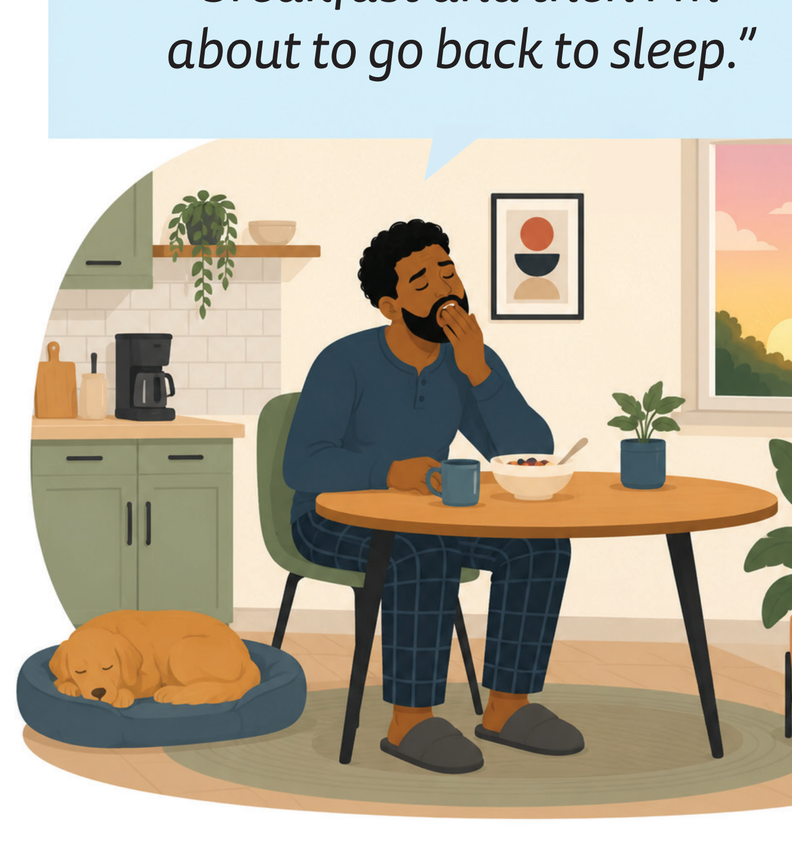

Patient voices on desired treatment outcomes

"Yeah, I mean there's been very few times in my entire life I've ever woken feeling refreshed. It is a wonderful feeling when you do get to feel it. It would be nice to have on a regular basis."

"I've only had a few times in my life and I would give anything to on a daily basis to feel refreshed."

"We want to be normal. If the sodium oxybate will help you sleep deeply, then maybe that's a good thing."

FIGURE 7. Desired Treatment Outcomes in ON-SXB-Experienced PWN

Patient voices on desired treatment outcomes

"I just wanted to sleep through the night and actually wake up feeling refreshed and not wake up feeling like I hadn't slept in two weeks."

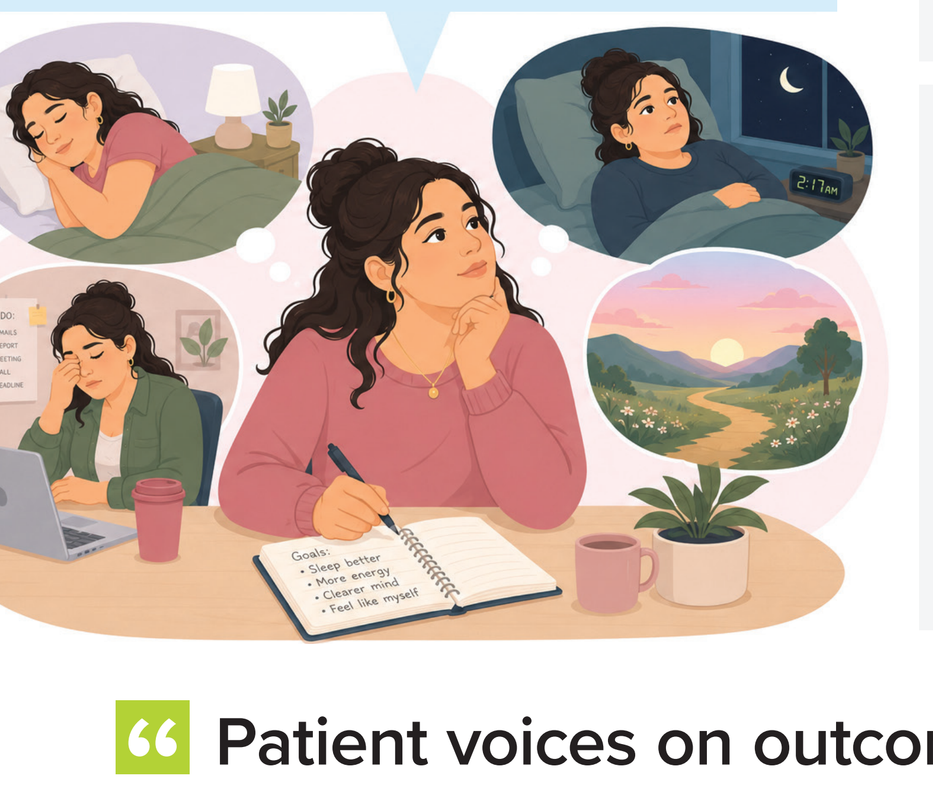

"I just wanted to feel like I had slept. It didn't matter how many hours I slept. I just wanted to sleep. The one thing that a lot of people take for granted and when you don't have it, it is huge."

"I wanted to sleep and then stay asleep long enough to feel refreshed. Waking up after every two minutes is not fun. It's not fun. It makes you feel that you can't do your job properly. Your mind works too slowly because it doesn't rest enough. And also the part about dreaming, the vivid dreaming, I wanted to improve my mental state."

Patient voices on outcomes achieved

"When I wake up in the morning, I'm good to go. I still get that afternoon slump and you know what? I just have to close my eyes. But it's much better than it used to be. So, I'm thankful for that."

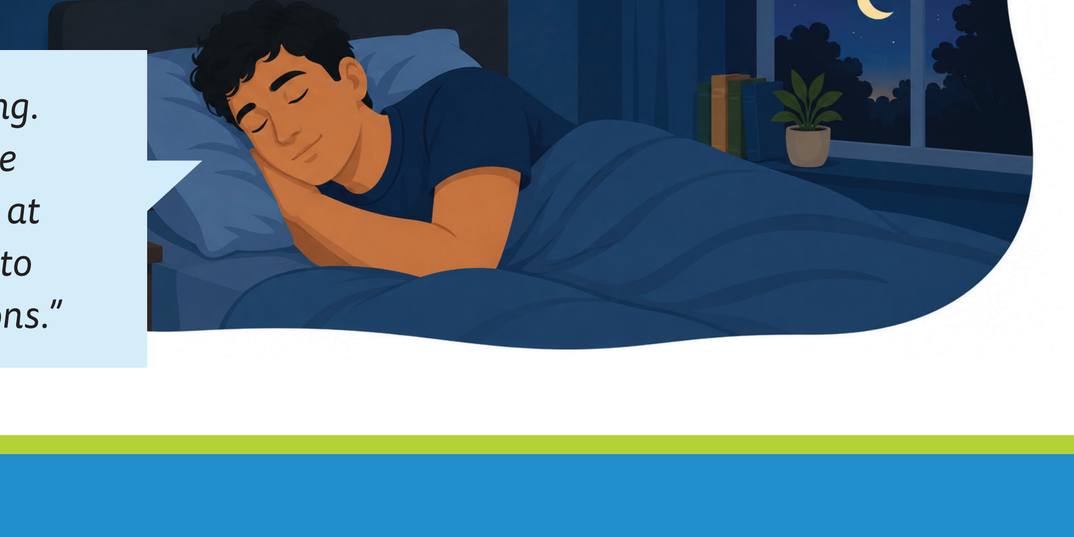

"A benefit is staying asleep for long. Right now, I get a consistent five hours, at least five hours of sleep at night compared to the one hour to half an hour for the last medications."

PRESENTED AT SLEEP 2026 • JUNE 14-17, 2026 • BALTIMORE, MD, USA

THESE MATERIALS ARE FOR PERSONAL USE ONLY AND MAY NOT BE REPRODUCED WITHOUT PERMISSION OF ALKERMES. FOR PERMISSION, CONTACT USMEDINFO@ALKERMES.COM