Real-World Patient Experience With Ruxolitinib Cream in Vitiligo

Swapna Ambati,[1] Dan Sturm,[1] Chris O'Brien,[2] Beth Schneider[2]

[1]Incyte Corporation, Wilmington, DE, USA; [2]MyHealthTeam, San Francisco, CA, USA

Presented at the
Fall Clinical Dermatology Conference
Las Vegas, NV, USA • October 23–26, 2025

Objective

- To assess real-world experiences of people with vitiligo who used ruxolitinib cream following FDA approval

Conclusions

- **Repigmentation in vitiligo often requires prolonged treatment, making sustained ruxolitinib cream use essential for optimal outcomes**
- **Patient-reported real-world experience highlights that ruxolitinib cream is well tolerated and associated with repigmentation rates that increase with longer treatment duration**
- **To help optimize repigmentation, ruxolitinib cream persistence may be further supported through patient education and clear treatment expectations**

Introduction

- Vitiligo is a chronic, inflammatory autoimmune disease that causes progressive skin depigmentation[1]
- People with vitiligo experience significant quality-of-life, psychiatric, and psychosocial burden[2]
- Ruxolitinib (selective JAK1/JAK2 inhibitor) cream is the first FDA-approved therapy for repigmentation of NSV in patients aged ≥12 years[3]

Methods

- Retrospective, cross-sectional online survey conducted from January 22 to February 17, 2025, through the MyVitiligoTeam platform, recruiting US adults with vitiligo aged ≥21 years
- The survey collected patient demographics, disease characteristics, treatment patterns, and HCP involvement in care

Results

Respondents

- Among the 309 respondents, 20.1% (n=62; **Table 1**) were current users of ruxolitinib cream, 11.3% (n=35) were former users, and 68.6% (n=212) were treatment-naive to ruxolitinib cream
 - Most current users were female and younger than 65 years
 - Current users most commonly had NSV and were managed by a dermatologist
- Despite one-third of current users having >10% BSA involvement, nearly all (91.9%) limited application to ≤10% of their BSA

Table 1. Characteristics of Current Ruxolitinib Cream Users

	Current users (n=62)
Age ≥65 y, %	37.1
Female, %	63.8
Race, %	
White	39.7
Black	20.7
Latino	20.7
Asian/Pacific Islander	5.2
Other	10.4
Prefer not to say	5.2
Vitiligo type, %	
NSV or generalized	71.0
Universal	6.5
Segmental	6.5
Not sure	16.1
HCP treating vitiligo, %	
Dermatologist	90.3
Primary care	6.5
Other	3.2
None	0
BSA, %	
≤10%	66.2
>10%	33.8

Treatment Patterns

- Among current users, ruxolitinib cream was most commonly applied to visible body areas, including the face, hands, neck, legs, and arms (**Figure 1**)
- Most current users applied ruxolitinib cream twice daily (74.2%) and more than half (56.5%) continued to treat their vitiligo for over 6 months (**Figure 2**)

Effectiveness

- Most current users (58.1%) reported experiencing repigmentation; rates of improvement increased with longer ruxolitinib cream treatment duration (**Figure 3**)

Safety

- Adverse events were reported by 40.3% of current users, most commonly application site itching (16.1%), headache (11.3%), and application site acne (11.3%)

Treatment Satisfaction

- Ruxolitinib cream treatment was well tolerated, with most current users likely to continue treatment (**Figure 4**)

Figure 1. Areas Where Current Users (n=62) Applied Ruxolitinib Cream*

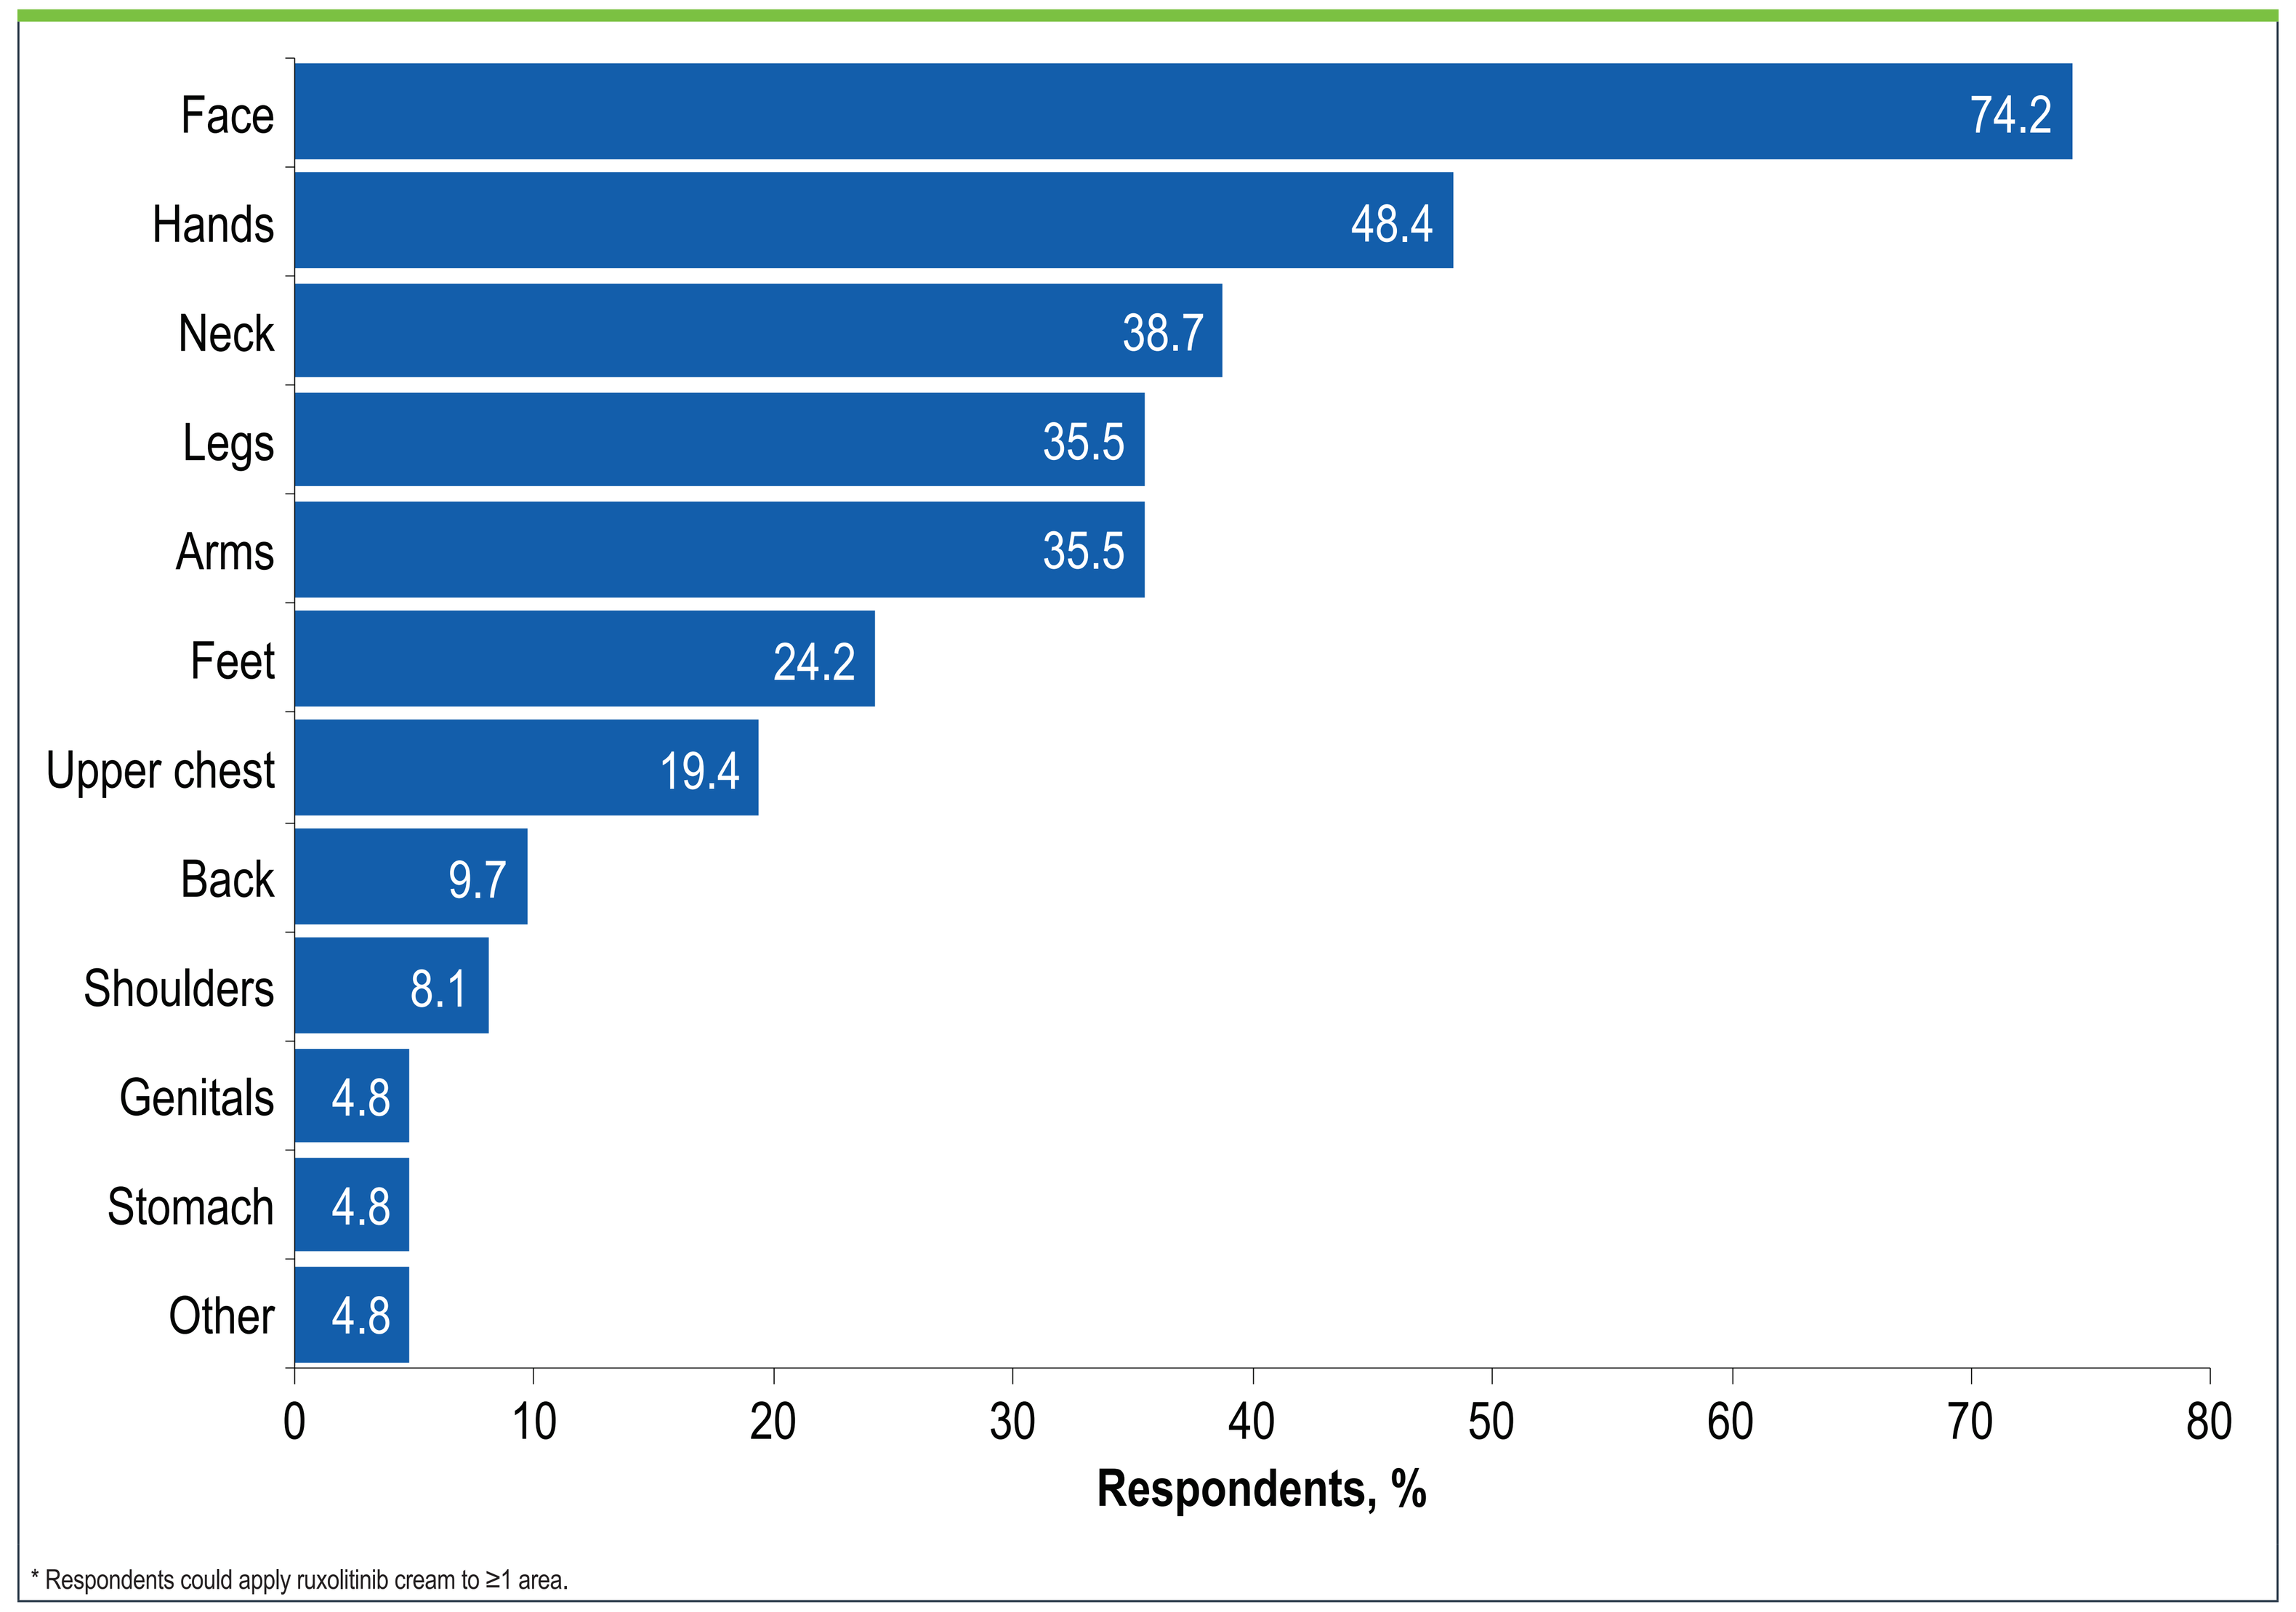
Face 74.2
Hands 48.4
Neck 38.7
Legs 35.5
Arms 35.5
Feet 24.2
Upper chest 19.4
Back 9.7
Shoulders 8.1
Genitals 4.8
Stomach 4.8
Other 4.8
0 10 20 30 40 50 60 70 80
Respondents, %

* Respondents could apply ruxolitinib cream to ≥1 area.

Figure 2. Frequency and Duration of Ruxolitinib Cream Use Among Current Users (n=62)

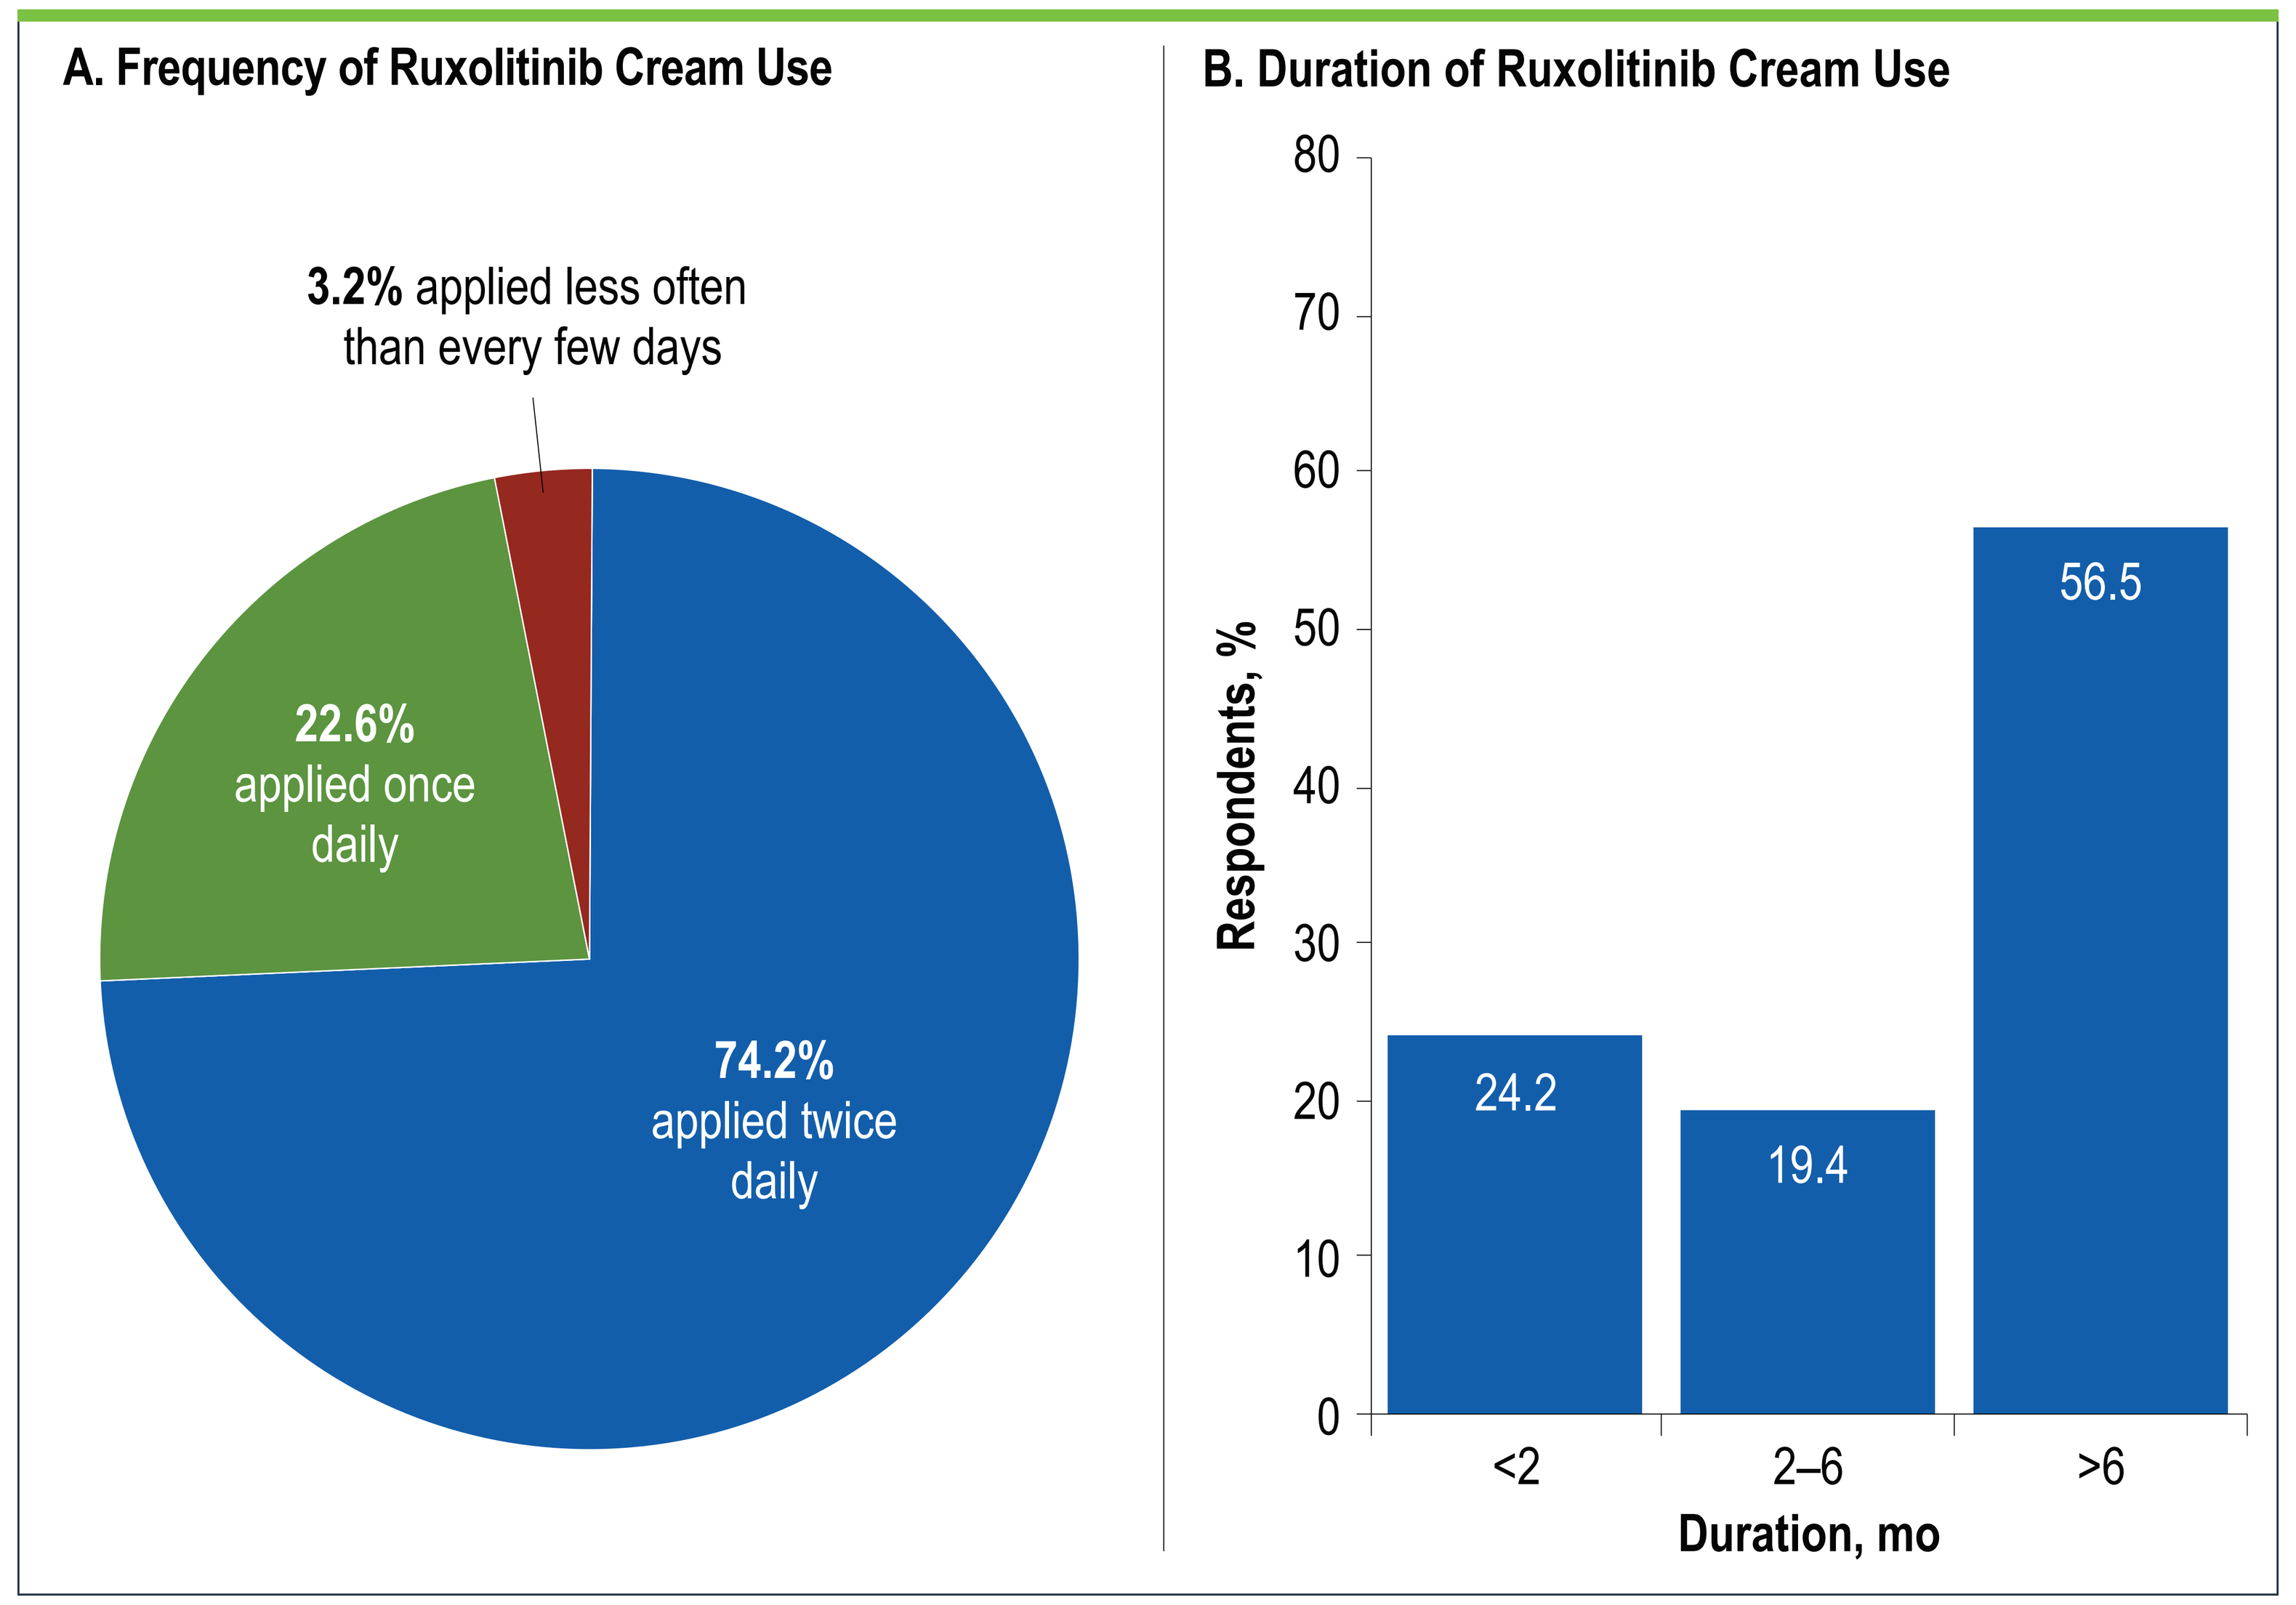
A. Frequency of Ruxolitinib Cream Use
3.2% applied less often than every few days
22.6% applied once daily
74.2% applied twice daily
B. Duration of Ruxolitinib Cream Use
Respondents, %
24.2
19.4
56.5
<2
2–6
>6
Duration, mo

Figure 3. Current Users (n=62) Experiencing Any Repigmentation by Ruxolitinib Cream Treatment Duration

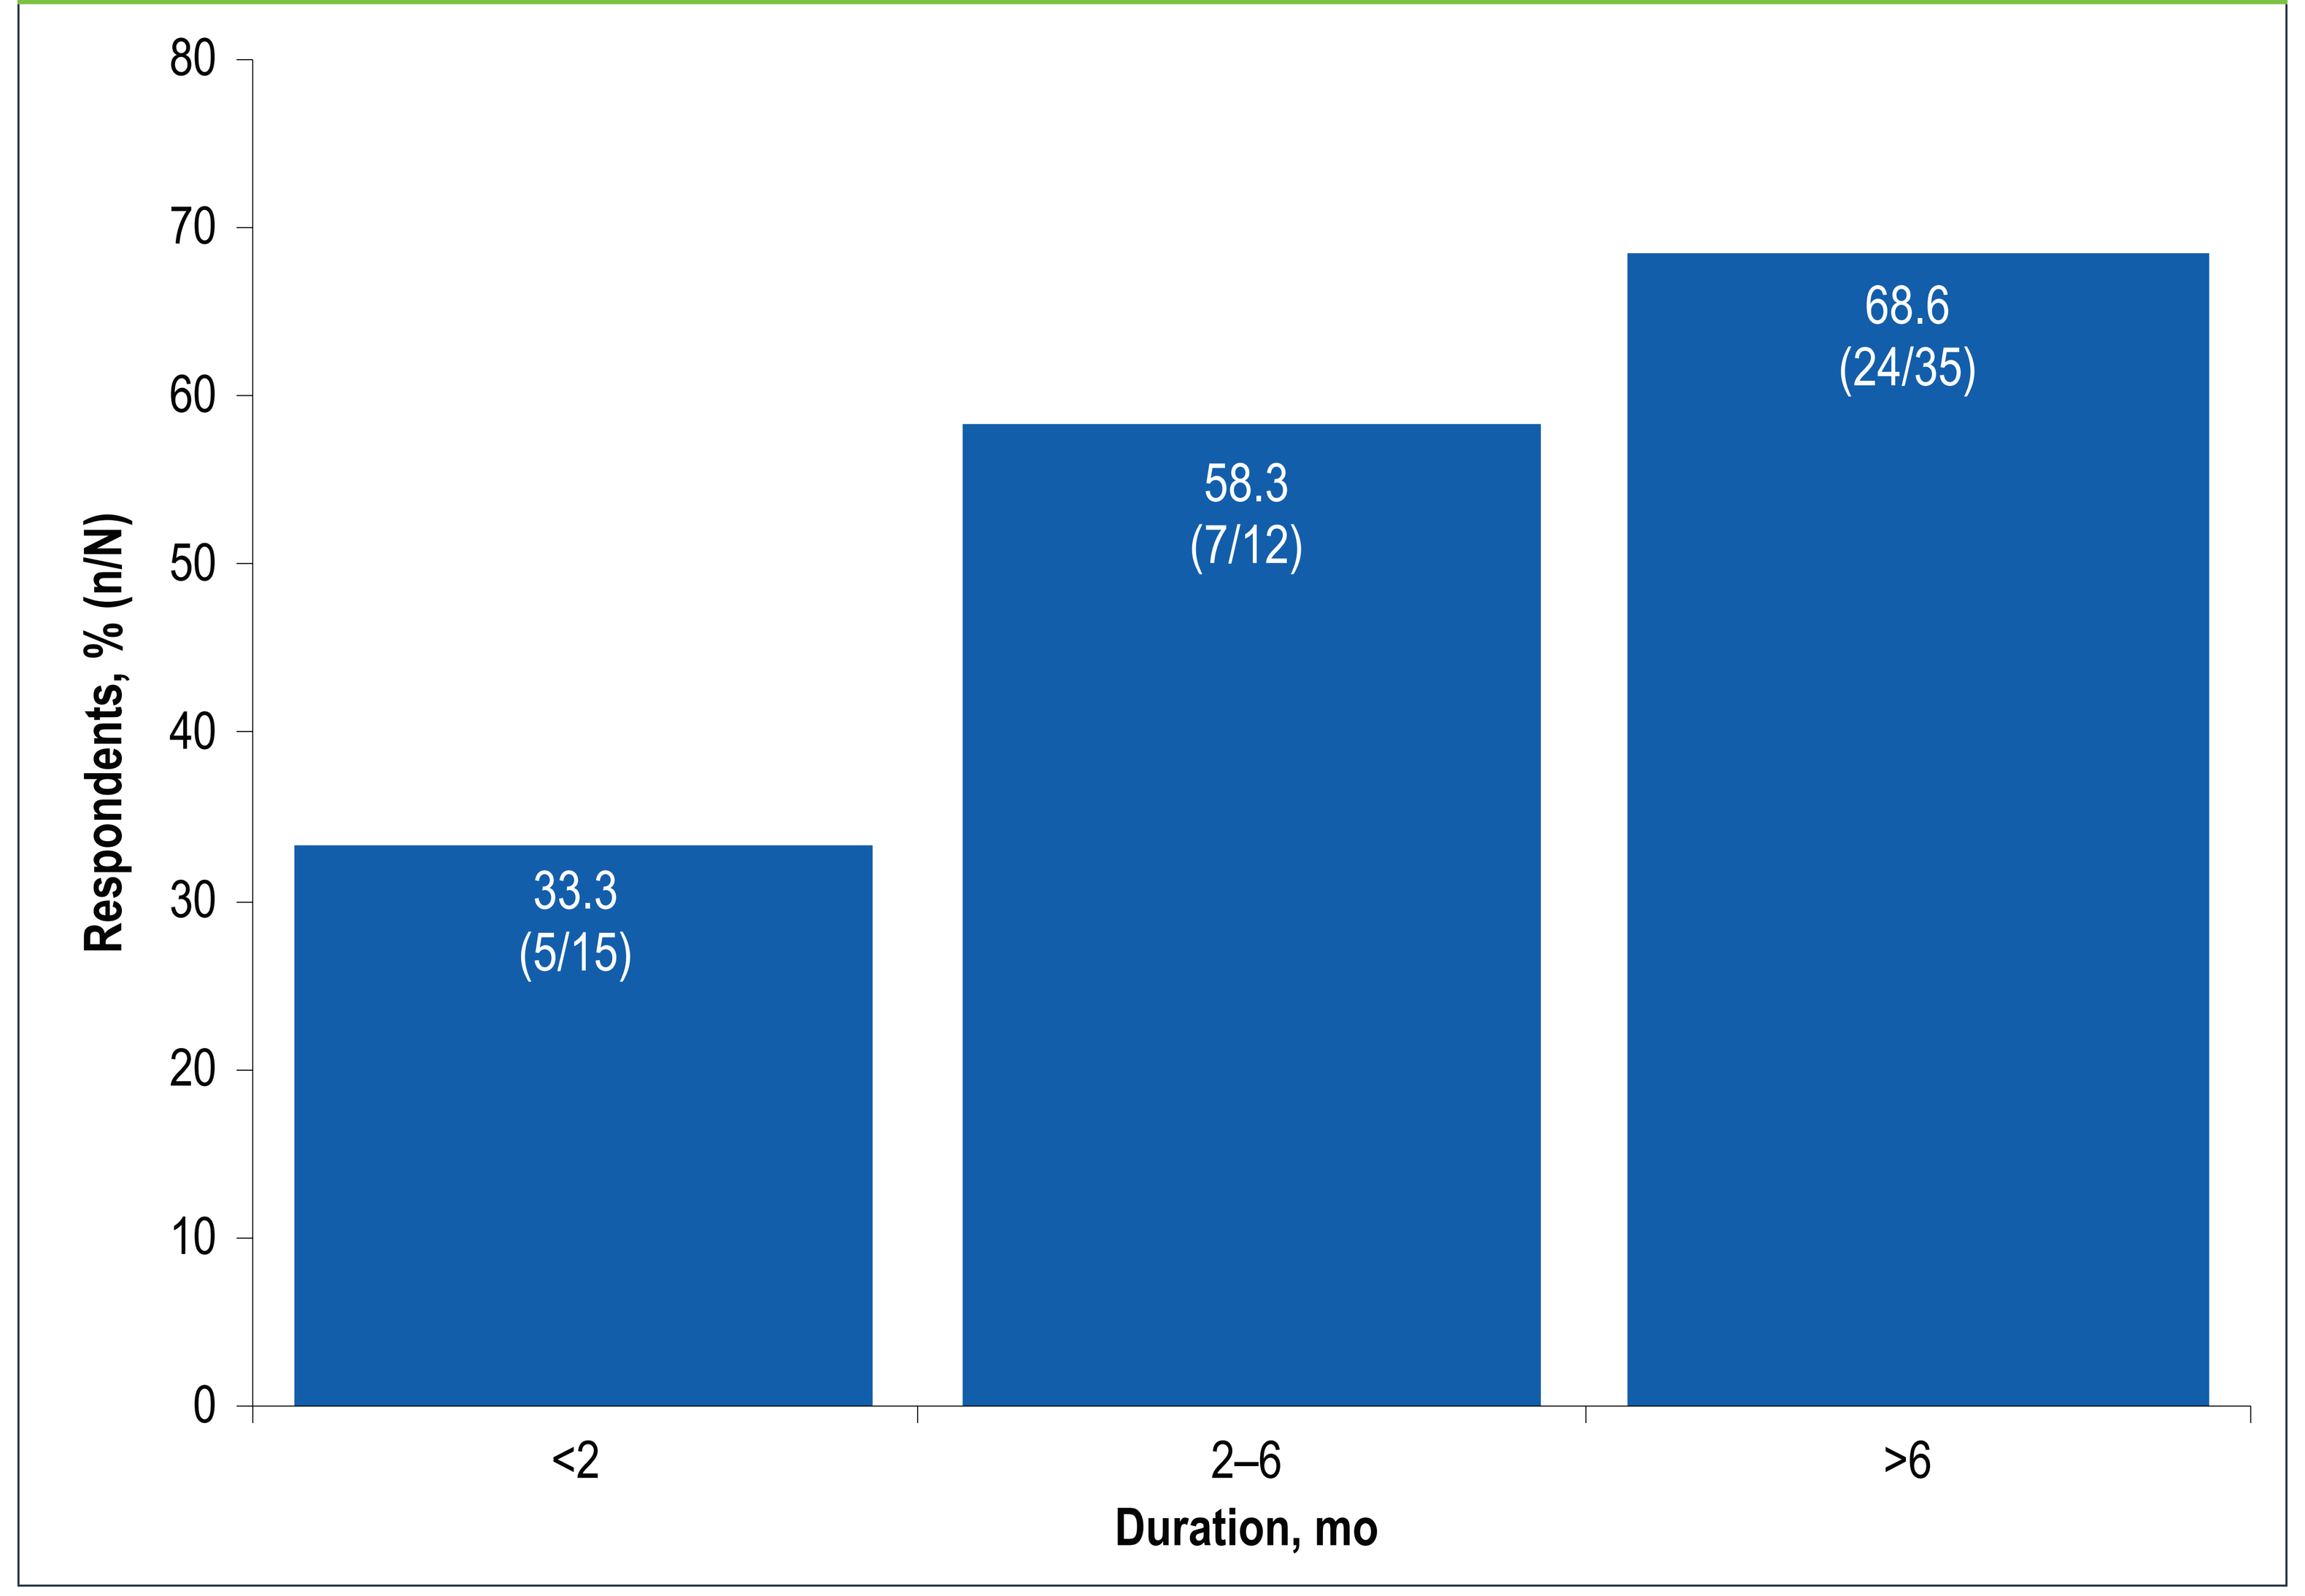
Respondents, % (n/N)
33.3 (5/15)
58.3 (7/12)
68.6 (24/35)
<2
2–6
>6
Duration, mo

Figure 4. Likelihood of Current Users (n=62) Remaining on Ruxolitinib Cream Treatment

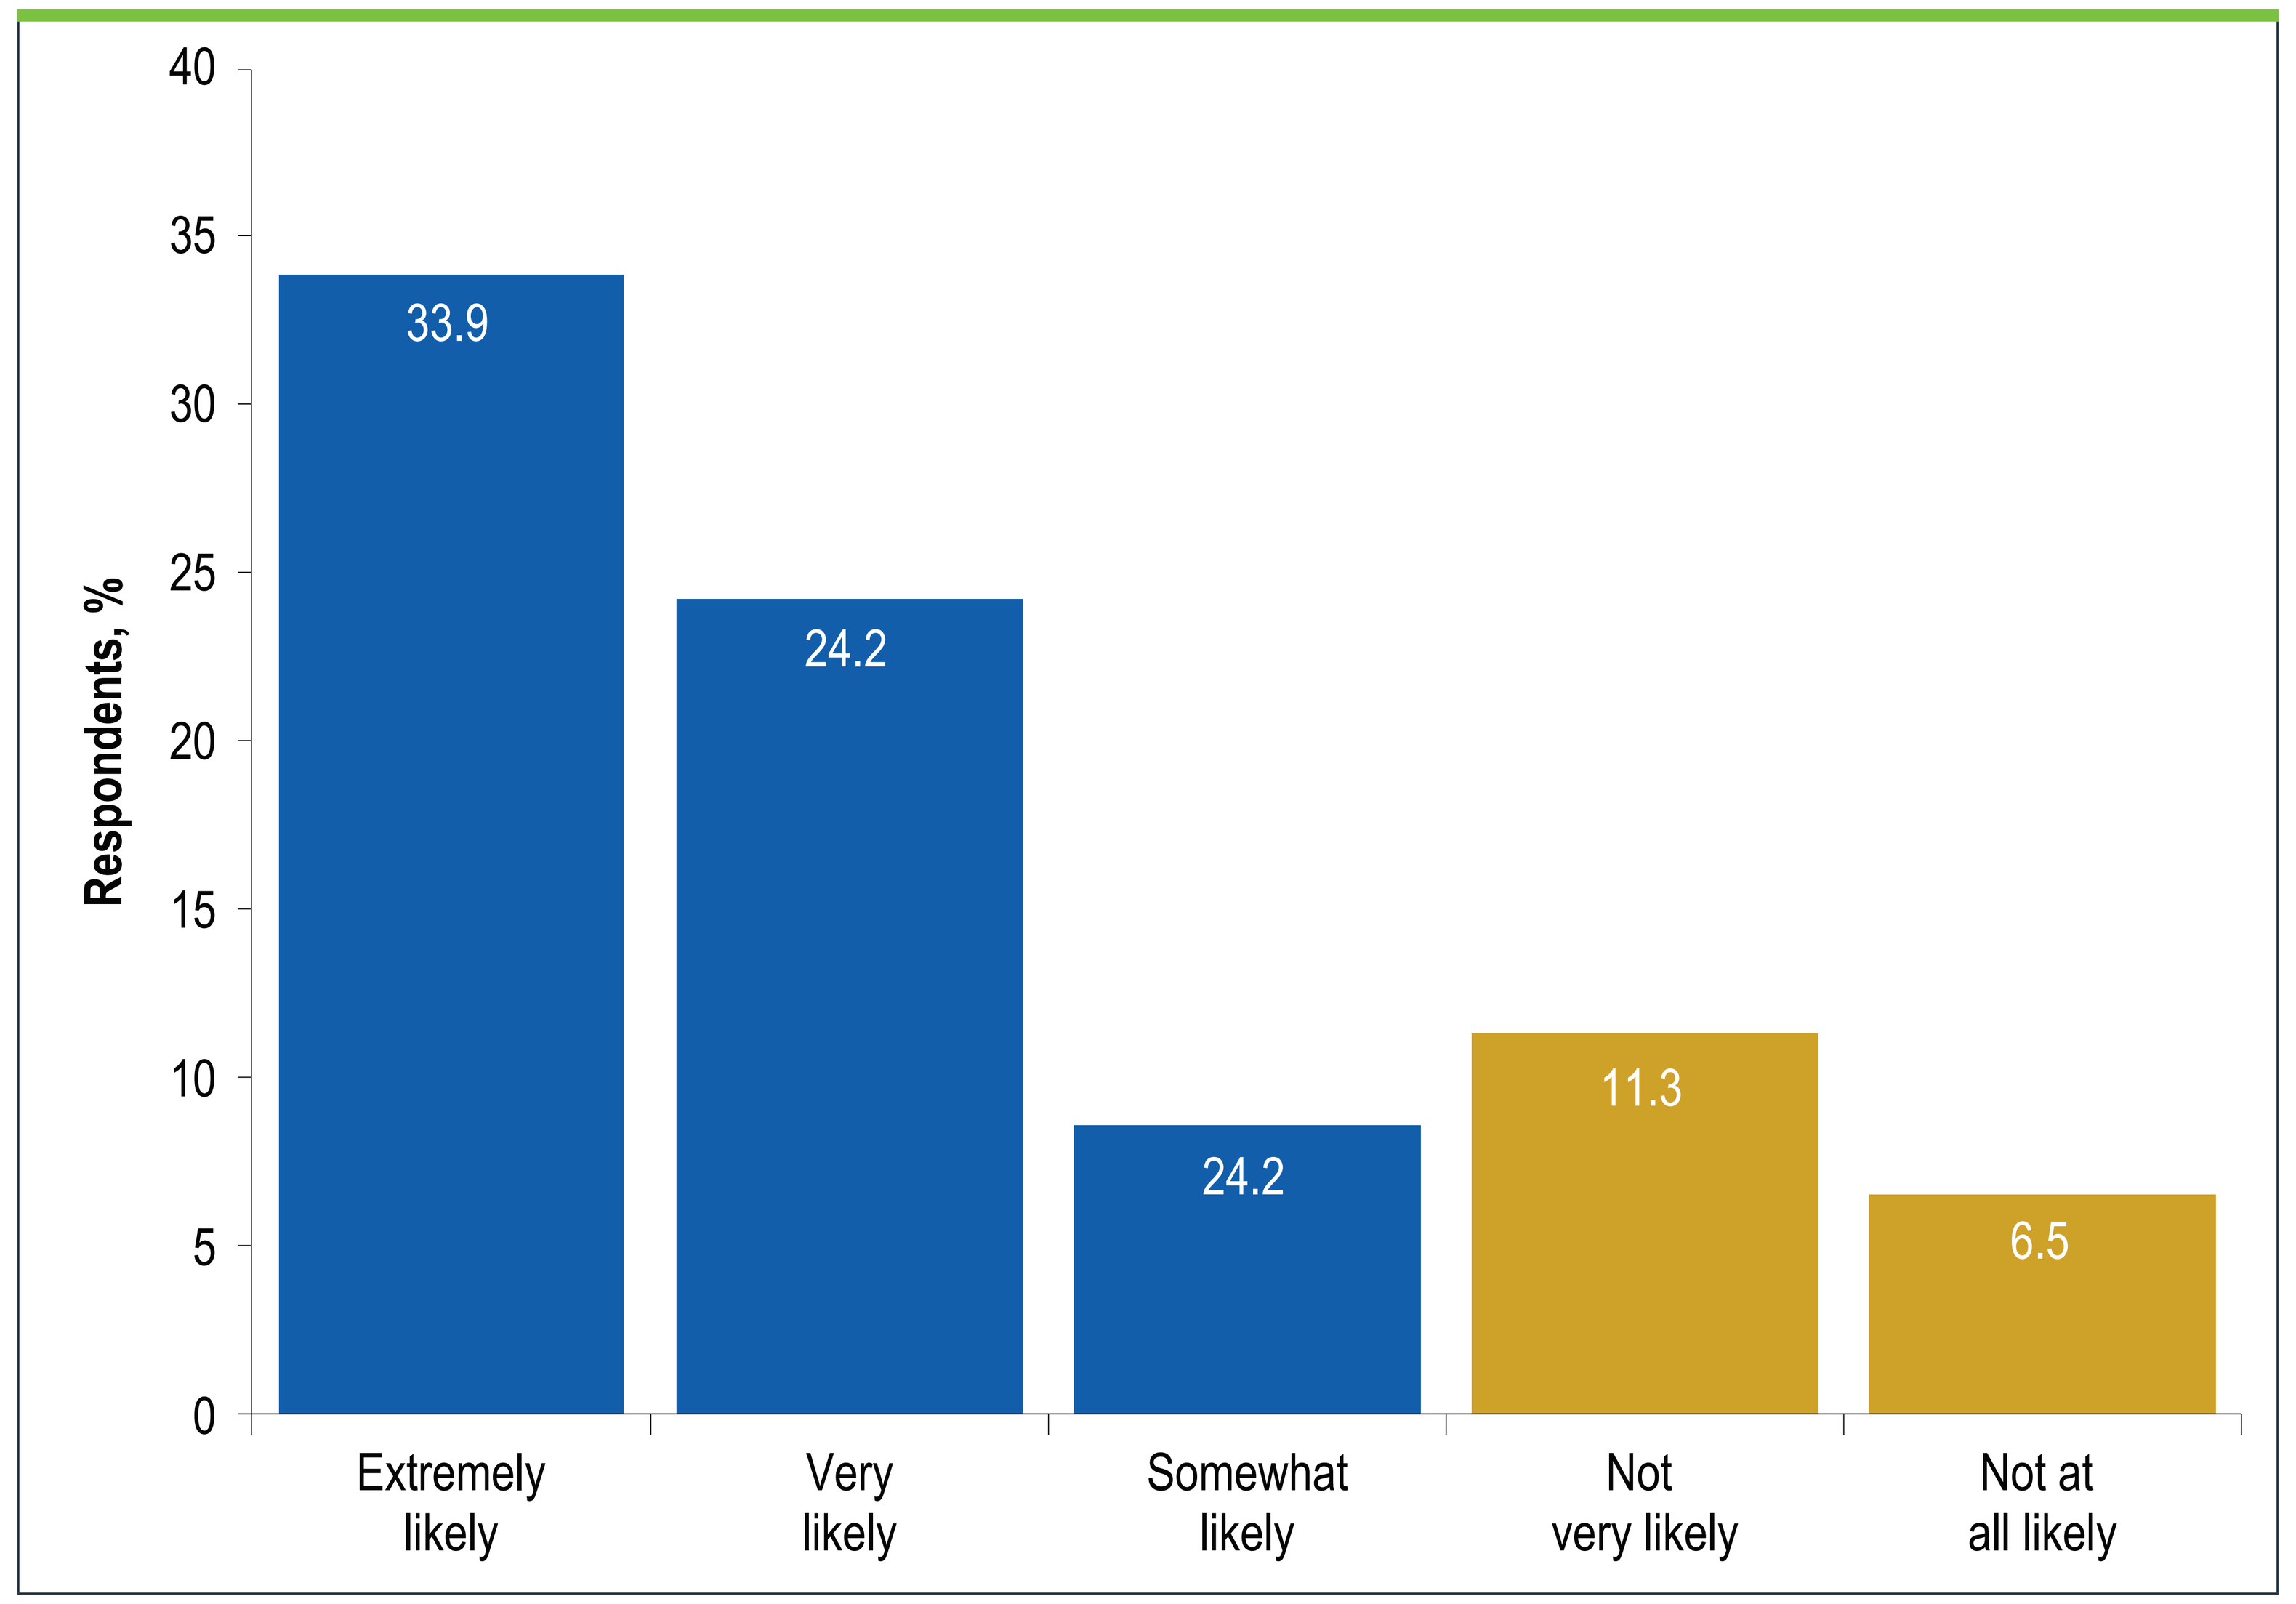
Respondents, %
33.9
24.2
24.2
11.3
6.5
Extremely likely
Very likely
Somewhat likely
Not very likely
Not at all likely

Abbreviations

BSA, body surface area; FDA, US Food and Drug Administration; HCP, healthcare provider; JAK, Janus kinase; NSV, nonsegmental vitiligo.

Disclosures

SA and **DS** are employees and shareholders of Incyte Corporation. **COB** and **BS** are employees of MyHealthTeam, a Swoop company, which received funding for conducting the survey on Incyte's behalf.

Acknowledgments

This study was sponsored by Incyte Corporation (Wilmington, DE). Medical writing assistance was provided by Miranda Tradewell, PhD, an employee of ICON (Blue Bell, PA), and was funded by Incyte.

References

1. Bergqvist C, Ezzedine K. *Dermatology*. 2020;236(6):571-592.
2. Rosmarin D, et al. *Dermatol Ther (Heidelb)*. 2024;14(6):1633-1647.
3. Seneschal J, et al. *J Eur Acad Dermatol Venereol*. 2023;37(11):2185-2195.

To download a copy of this poster, scan code.