Poster# 1904

Patient Reported Attitudes Towards Multiple Sclerosis Treatment

Authors: Dianna Cronin (MyHealthTeam) and Beth Schneider (MyHealthTeam)

CONCLUSIONS

Understanding the quality-of-life impact of MS along with patient desired outcomes can help neurologists develop the most effective approach to treating patients. This includes listening to patient concerns about getting on DMTs, helping them weigh the concerns against benefits and educating them on the importance of treatments to slow progression rather than first waiting for MS to worsen.

BACKGROUND

Obstacles exist for getting patients on disease modifying treatments (DMTs) to slow progression. Understanding patient concerns can enable neurologists to help patients overcome barriers.

OBJECTIVES

Research was undertaken to better understand awareness, usage, and attitudes towards DMTs.

DESIGN/METHOD

In January 2024, an online survey was sent to US members of MyMSTeam. In total, 1294 members completed the survey.

AAN 2025

RESULTS

Only 18% of respondents rated their overall MS health as excellent or good. **(Figure 1)**

Most felt MS impacts quality of life (82%), including physical limitations (78%). **(Figure 2)**

84% of these patients were taking non-DMT medications to address MS symptoms such as spasticity (43%), pain (40%), depression (38%), anxiety (32%), or fatigue (31%). **(Figure 3)**

A smaller number (60%) are currently on a DMT. An additional 24% had taken one or more DMTs previously but had stopped. **(Figure 4)**

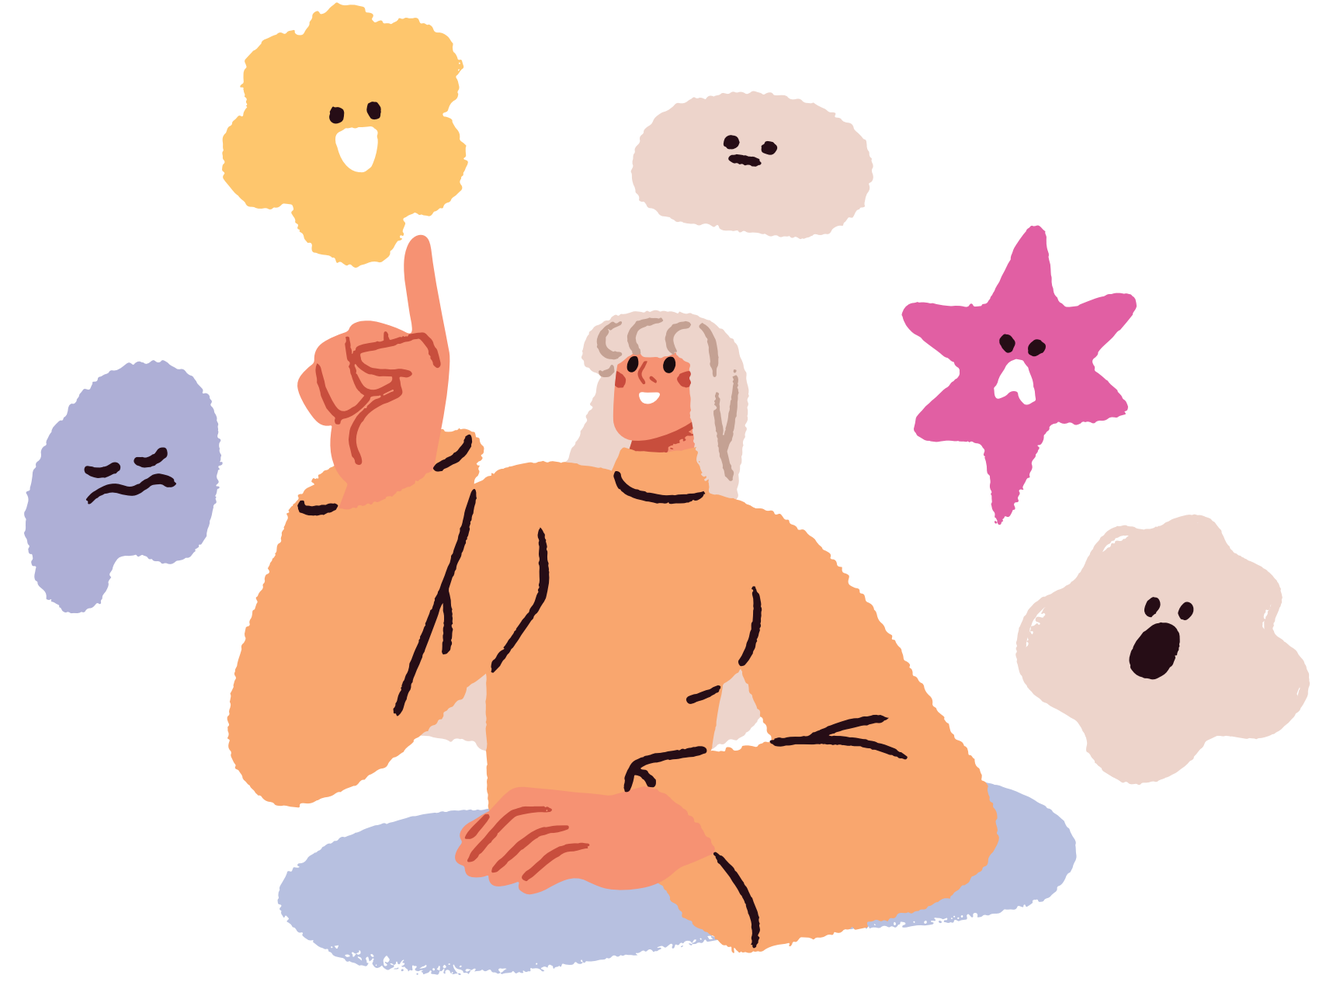

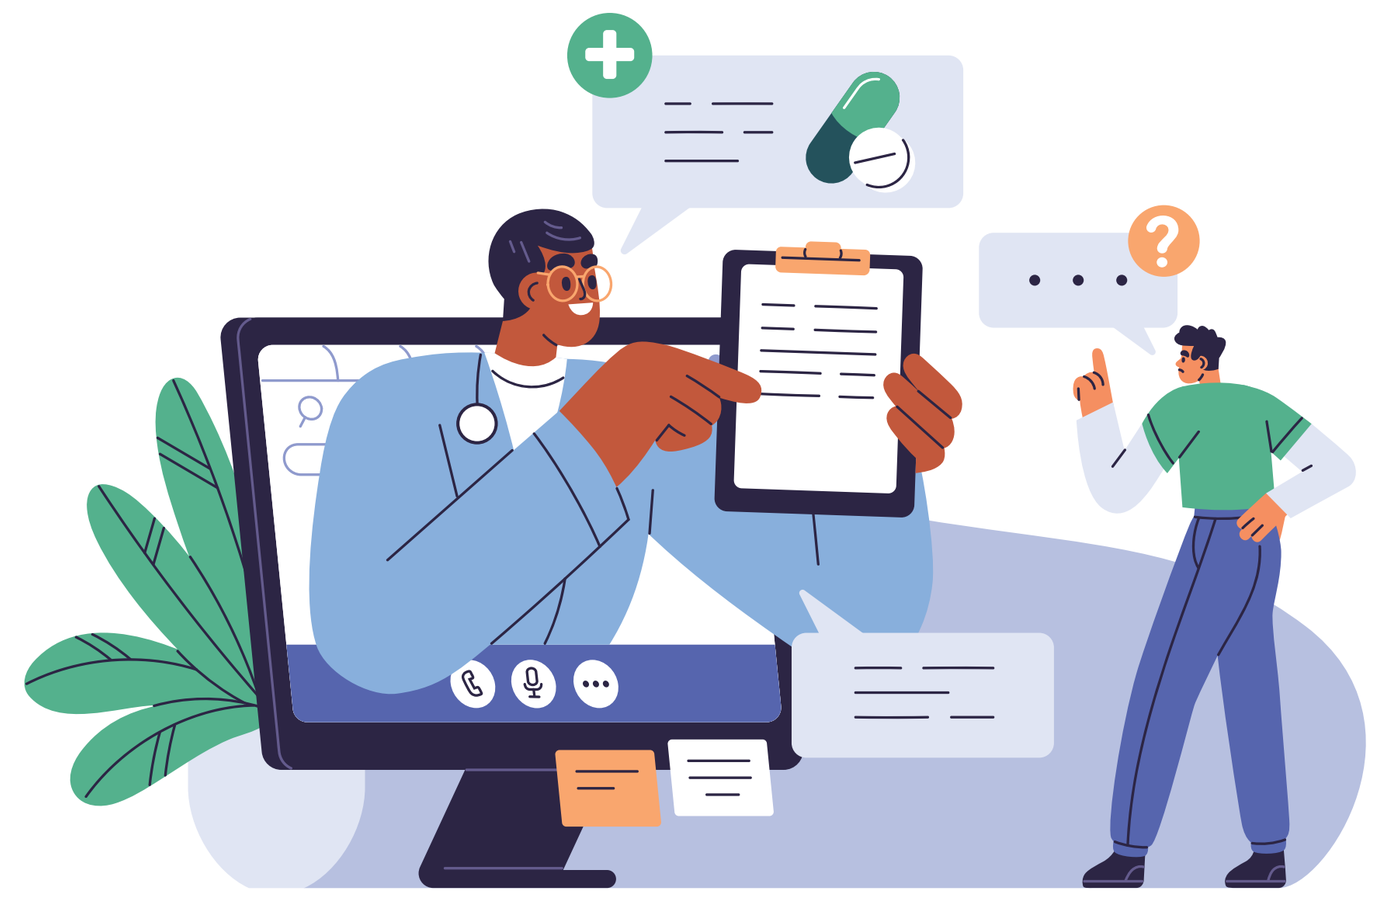

These abandoners might consider going back on a DMT if their doctor recommends (45%), MS progresses (43%), or symptoms worsen (42%). **(Figure 5)**

The neurologist was paramount in patient's decision to start the current DMT (79%). Perceived efficacy (61%), insurance approval (53%), and convenience (46%) also mattered. **(Figure 6)**

Current users are satisfied with their DMT (71%), feel it is effective (69%), and find it tolerable (86%). Benefits include slowed progression (68%), preventing new lesions (56%), a decrease in relapses (48%), and preventing new symptoms (42%). **(Figure 7)**

Benefits are consistent with desired patient outcomes of a DMT. **(Figure 8)**

Patients who had never taken a DMT were concerned about the potential safety or side effects (39%), insurance/cost hurdles (19%), or inconvenience (14%). This cohort was most likely to consider starting on a DMT if their neurologist strongly recommended it (36%), if safety concerns were allayed (35%), or if MS started to worsen (35%). **(Figure 9)**

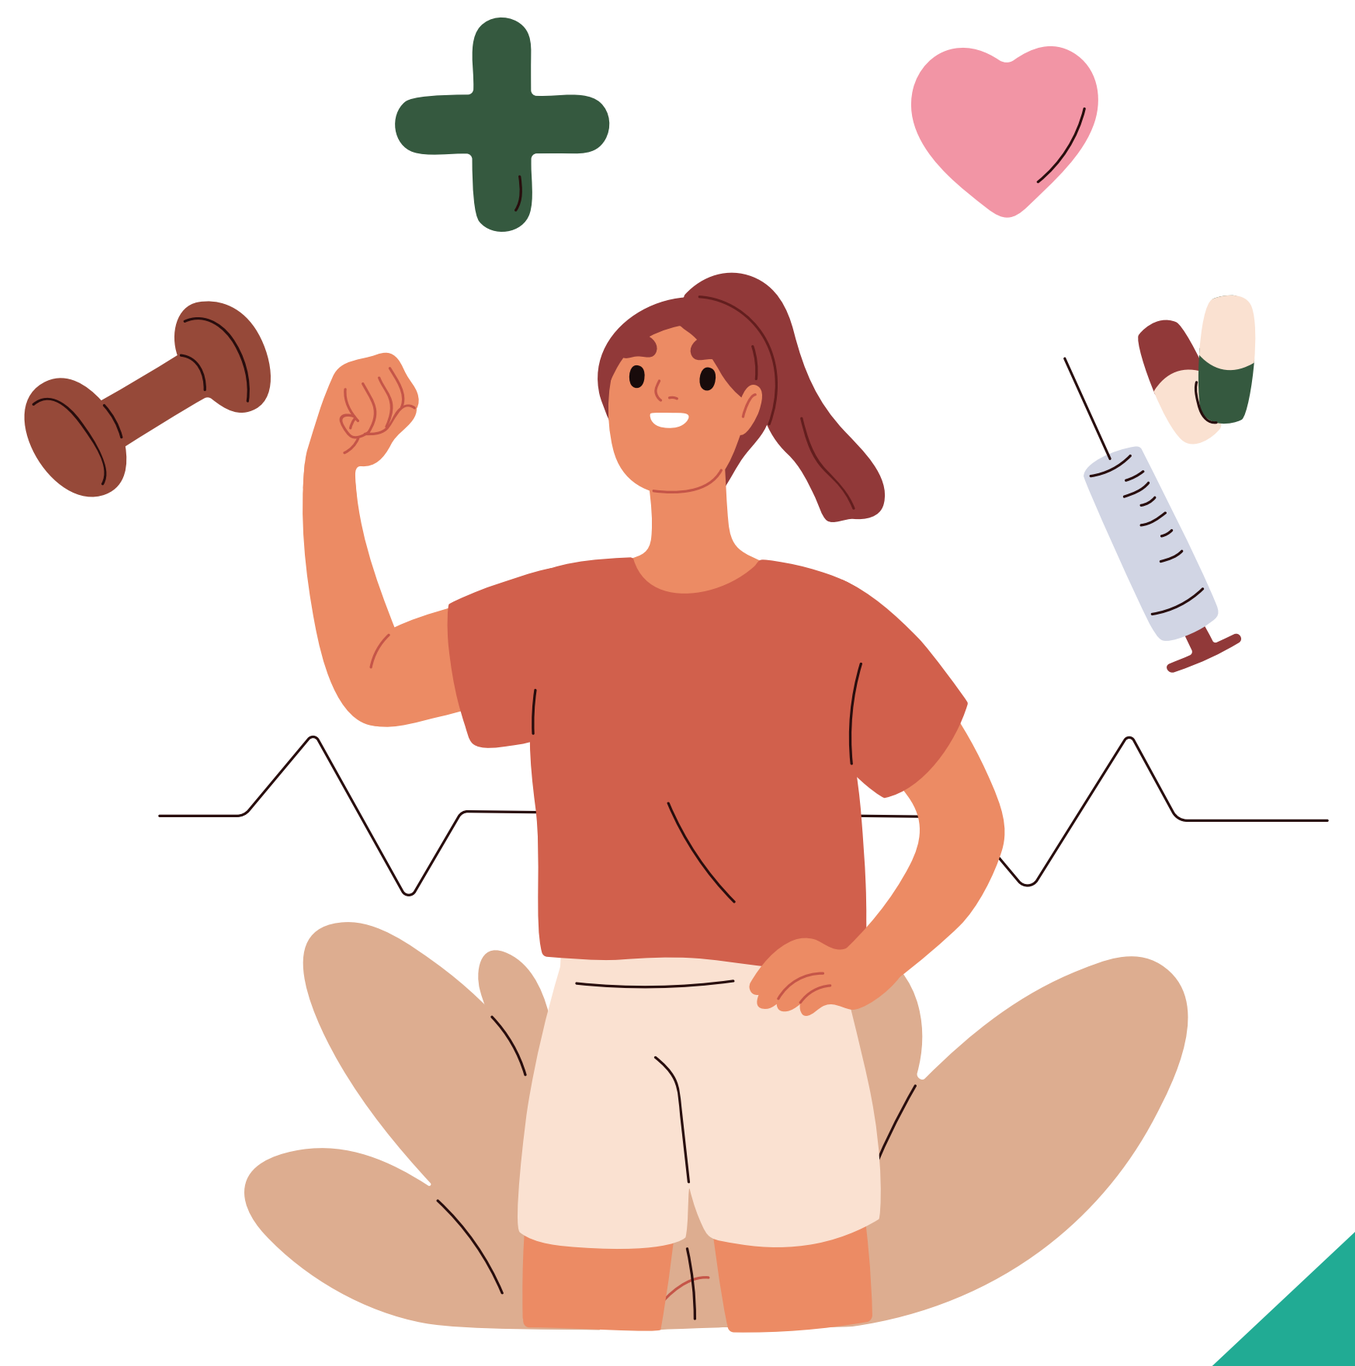

FIGURE 1. MS Related Health

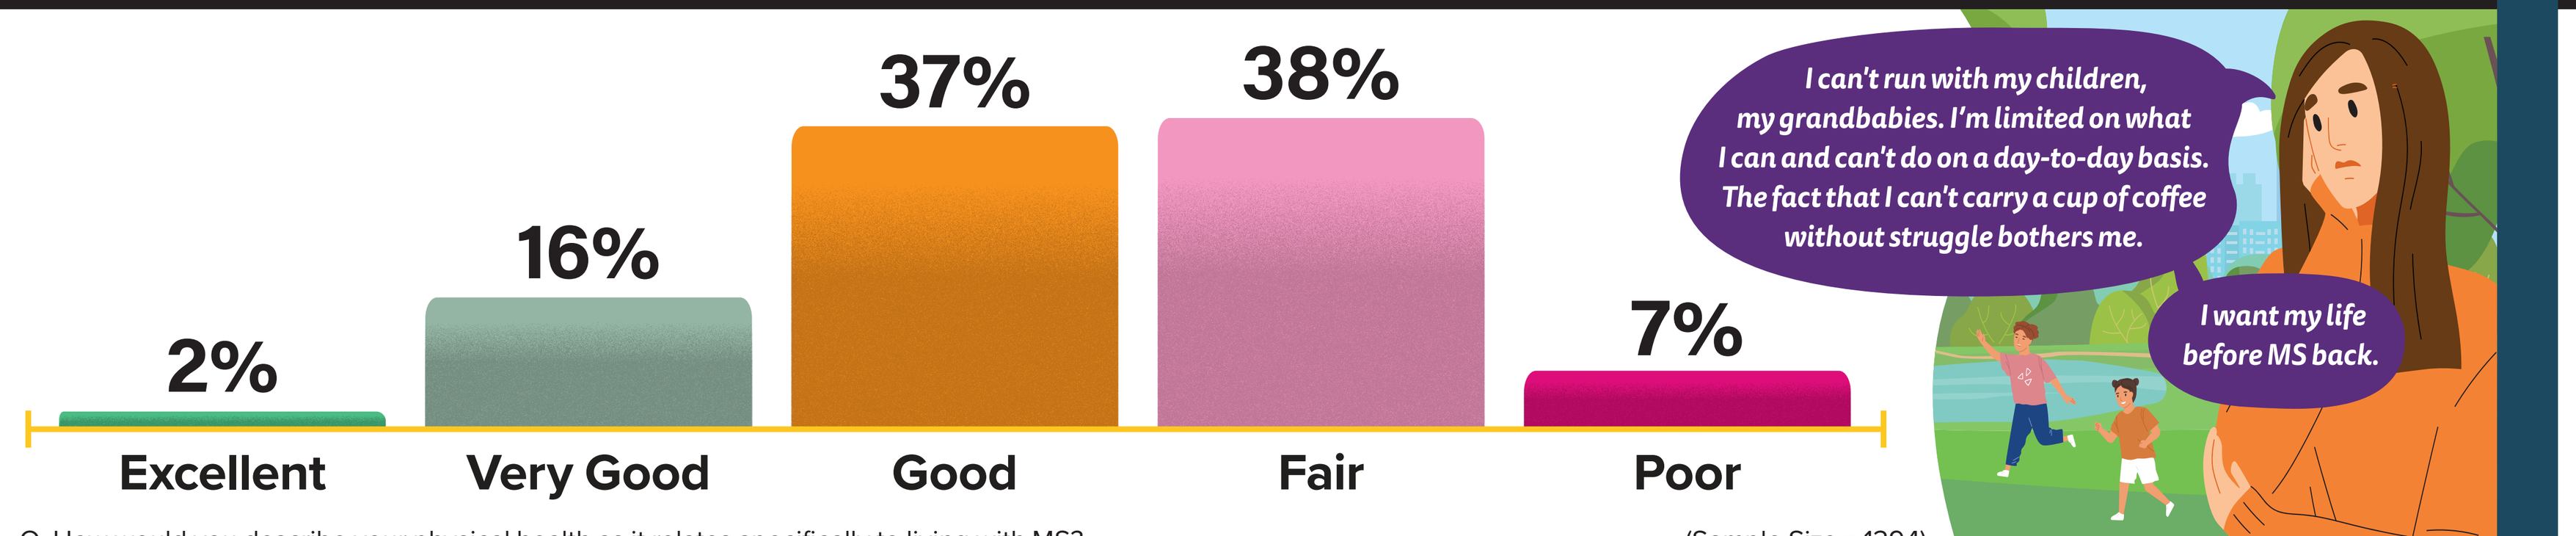
2%
16%
37%
38%
7%
Excellent
Very Good
Good
Fair
Poor
I can't run with my children, my grandbabies. I'm limited on what I can and can't do on a day-to-day basis. The fact that I can't carry a cup of coffee without struggle bothers me.
I want my life before MS back.

Q. How would you describe your physical health as it relates specifically to living with MS? (Sample Size = 1294)

FIGURE 2. Impact of MS on Quality of Life % Strongly/Somewhat Agreeing

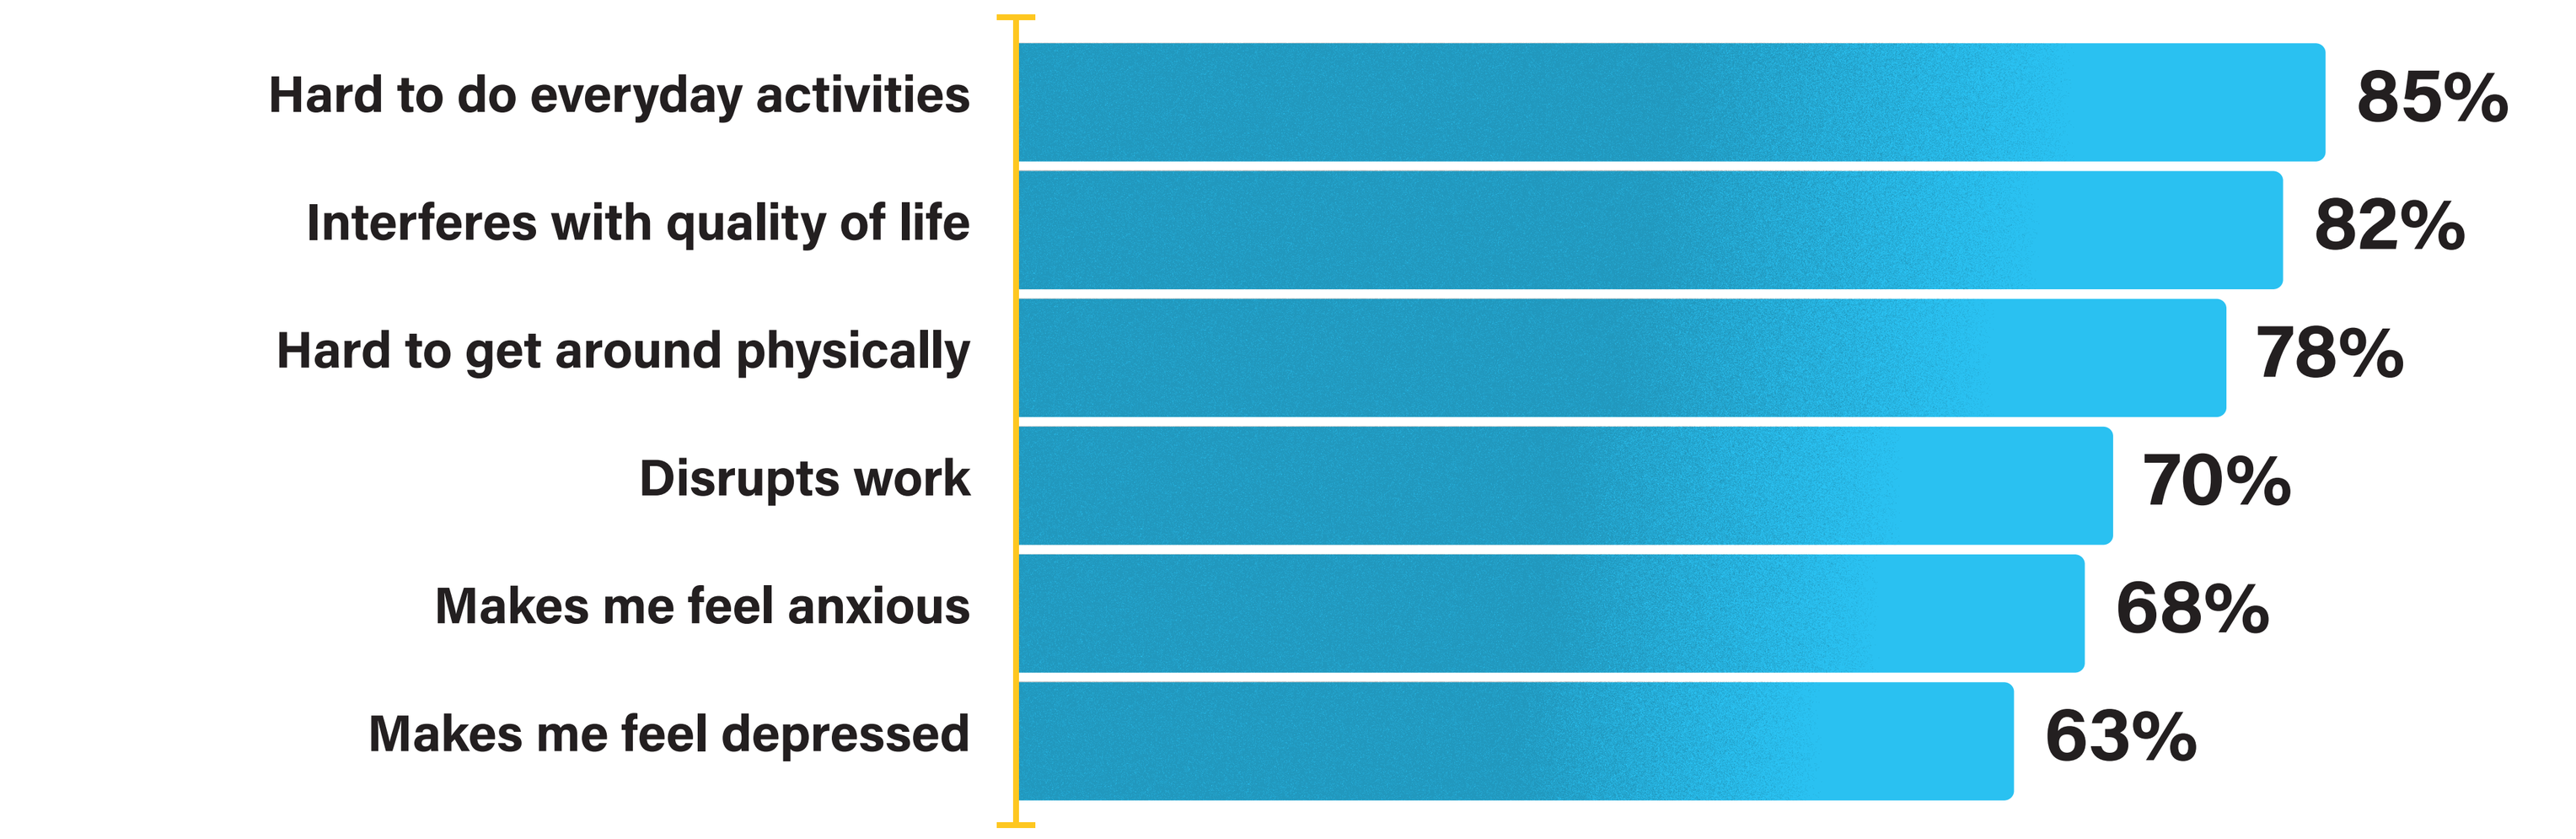
Hard to do everyday activities 85%
Interferes with quality of life 82%
Hard to get around physically 78%
Disrupts work 70%
Makes me feel anxious 68%
Makes me feel depressed 63%

Q. How much do you agree or disagree with the following statements? Having MS... (Sample Size = 1294)

FIGURE 3. MS Symptoms Requiring Medication

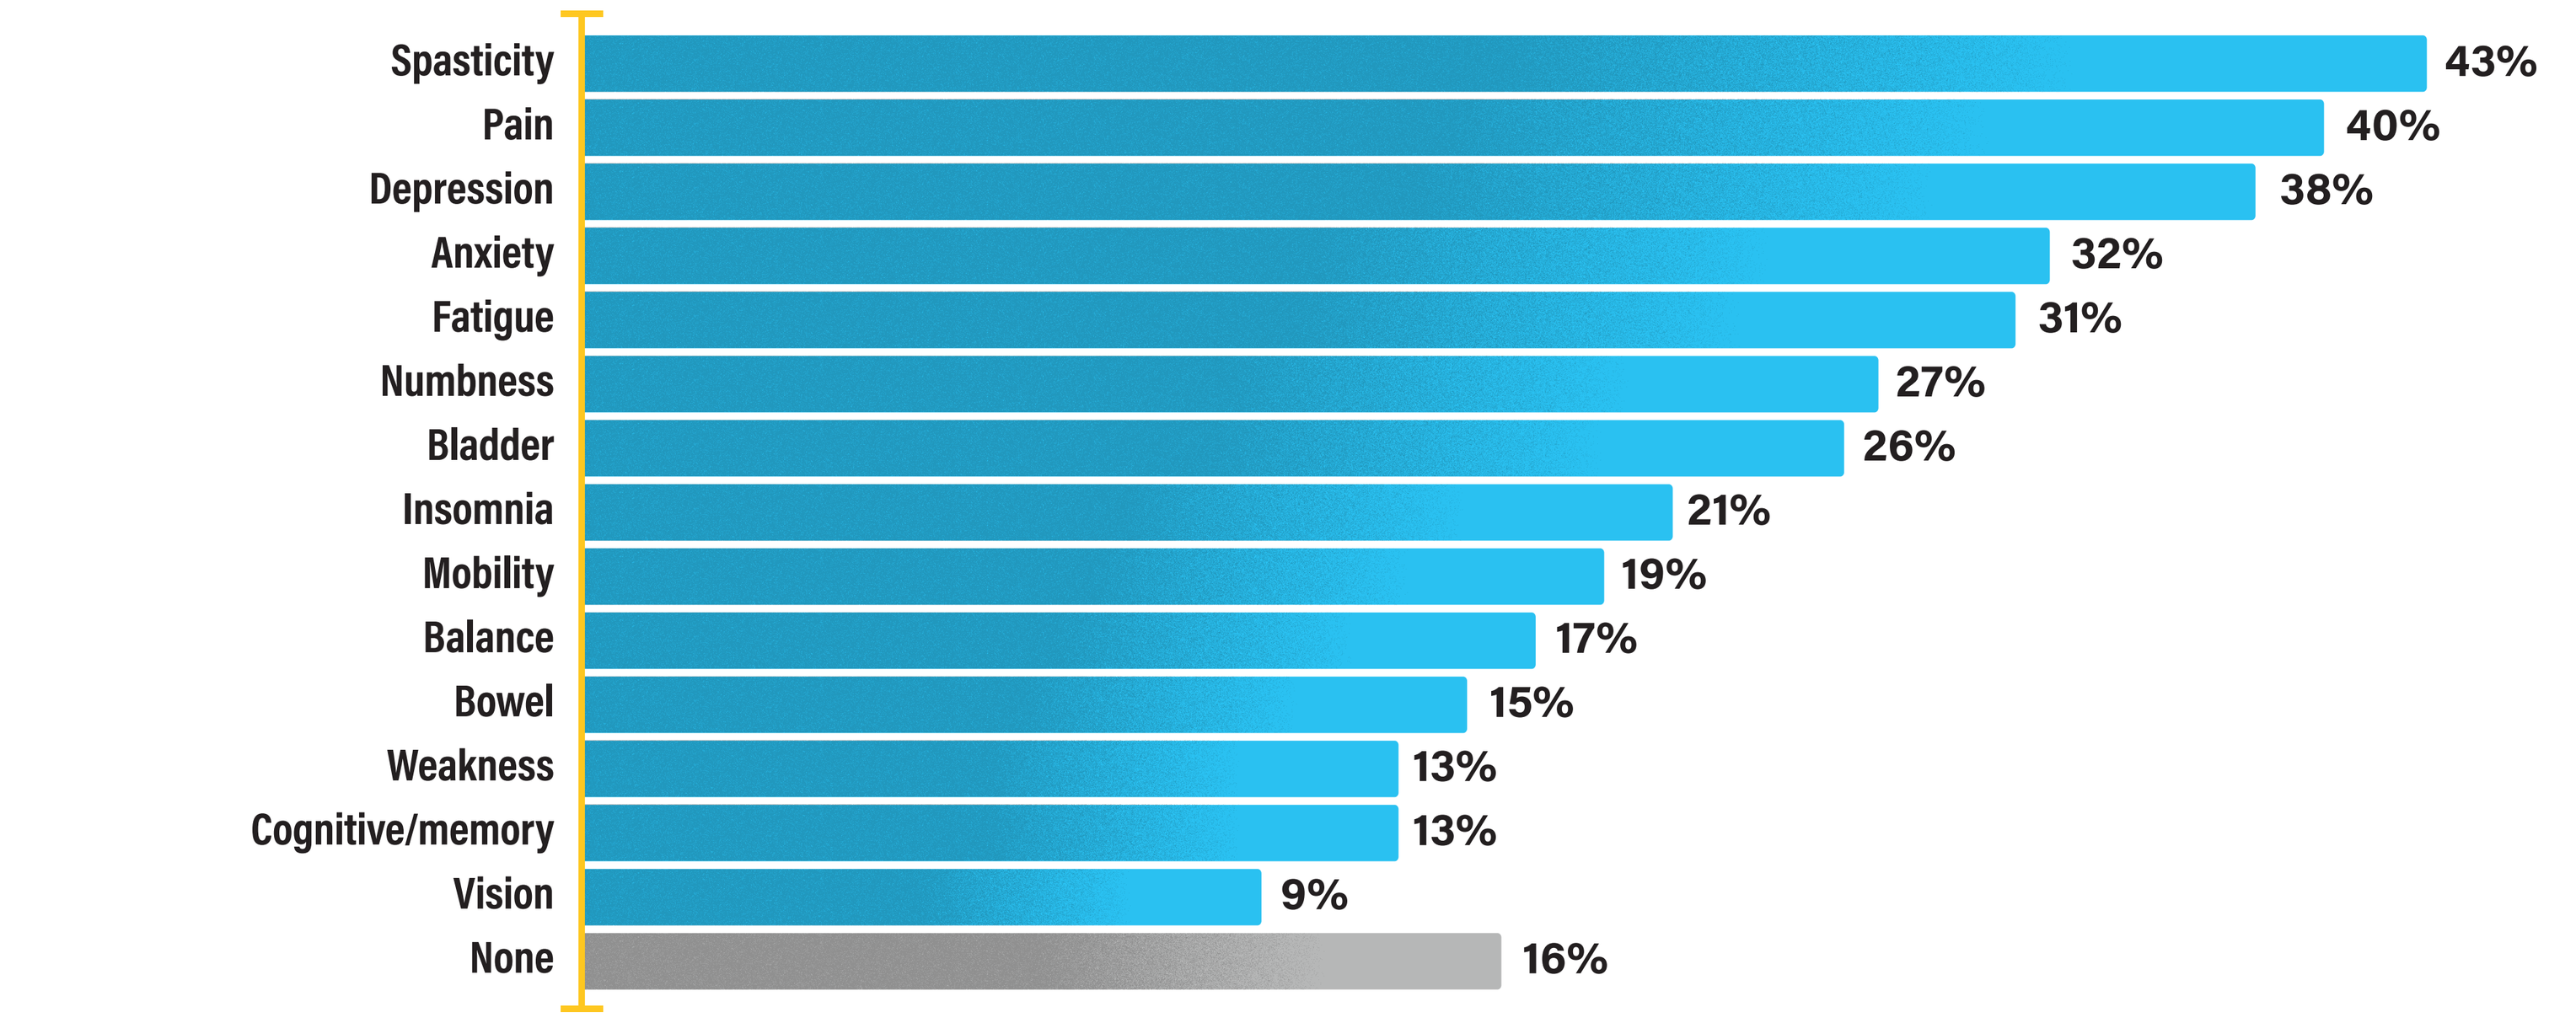
Spasticity 43%
Pain 40%
Depression 38%
Anxiety 32%
Fatigue 31%
Numbness 27%
Bladder 26%
Insomnia 21%
Mobility 19%
Balance 17%
Bowel 15%
Weakness 13%
Cognitive/memory 13%
Vision 9%
None 16%

Q. Apart from DMTs to slow progression, are you taking medications to address any of the following MS-related symptoms? Select all that apply. (Sample Size = 1294)

FIGURE 4. Experience With DMTs

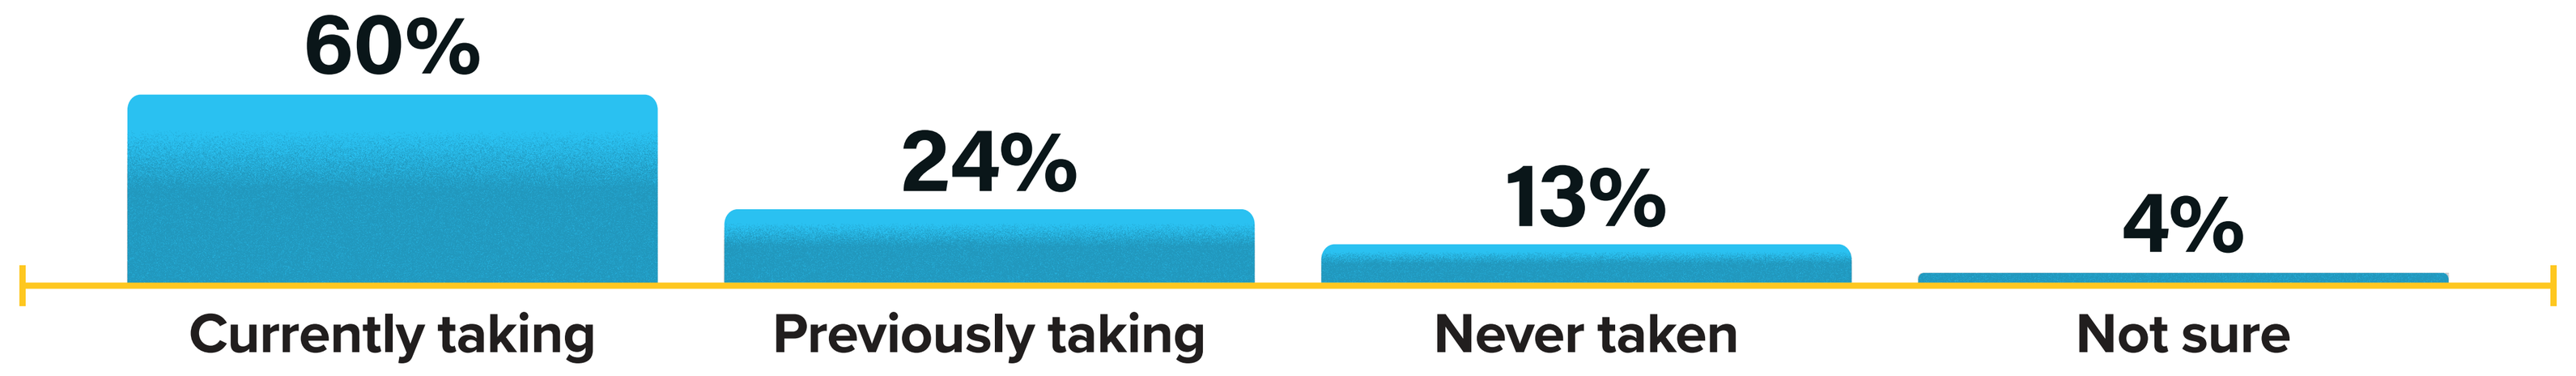
60%
24%
13%
4%
Currently taking
Previously taking
Never taken
Not sure

Q. Are you currently taking a disease modifying MS treatment, ("DMT") or have you ever taken one? (Sample Size = 1294)

FIGURE 5. Reasons Might Re-Start Using a DMT

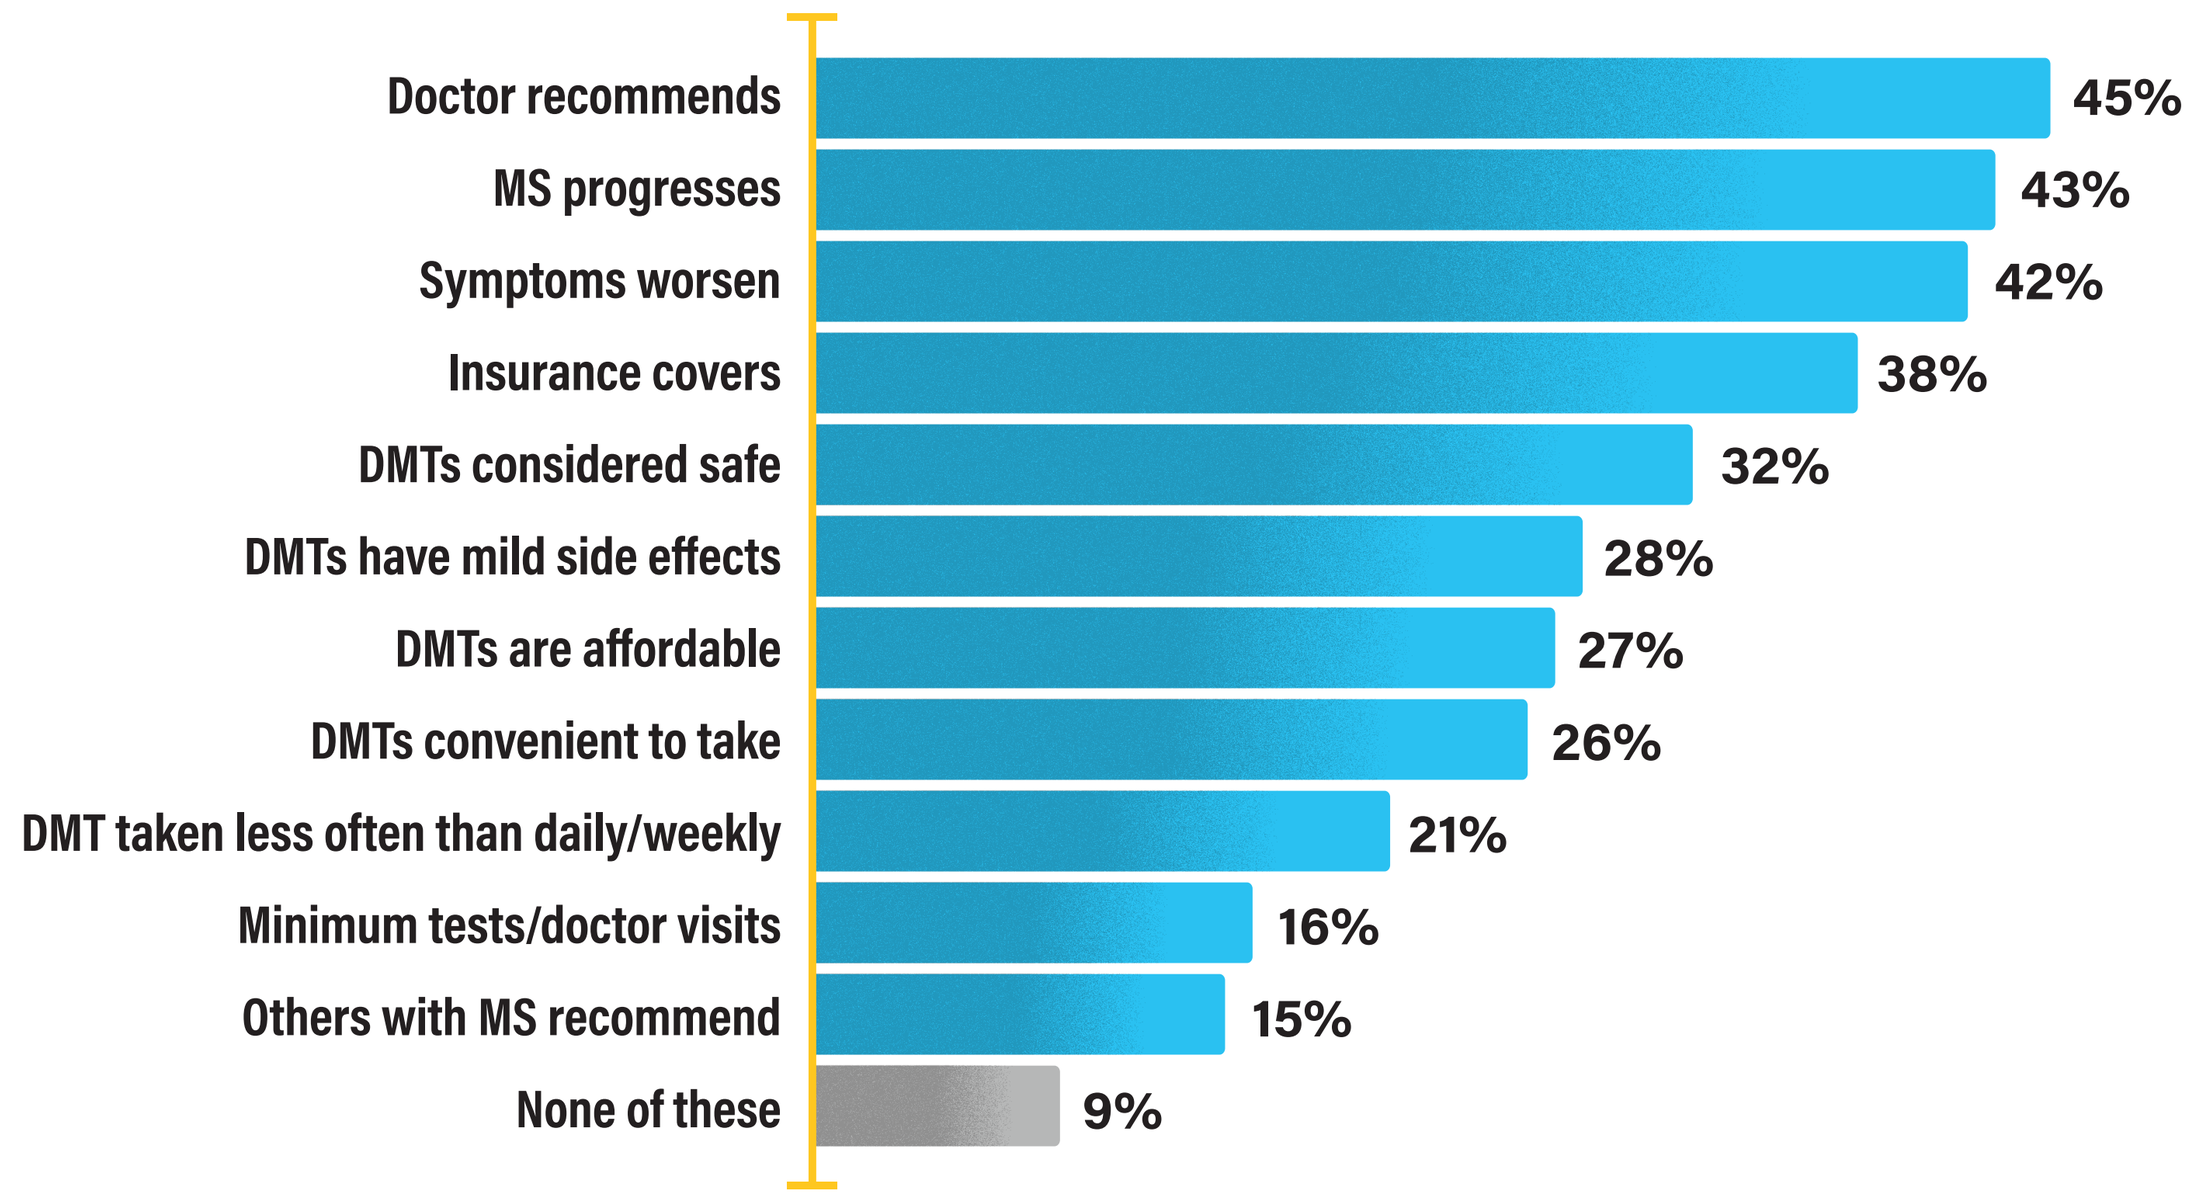
Doctor recommends 45%
MS progresses 43%
Symptoms worsen 42%
Insurance covers 38%
DMTs considered safe 32%
DMTs have mild side effects 28%
DMTs are affordable 27%
DMTs convenient to take 26%
DMT taken less often than daily/weekly 21%
Minimum tests/doctor visits 16%
Others with MS recommend 15%
None of these 9%

Q. Which, if any, of the following might be reasons why you would start taking prescription DMTs in the future? Select all that apply. (Asked of those previously on a DMT) (Sample Size = 311)

FIGURE 6. Why Started Current DMT

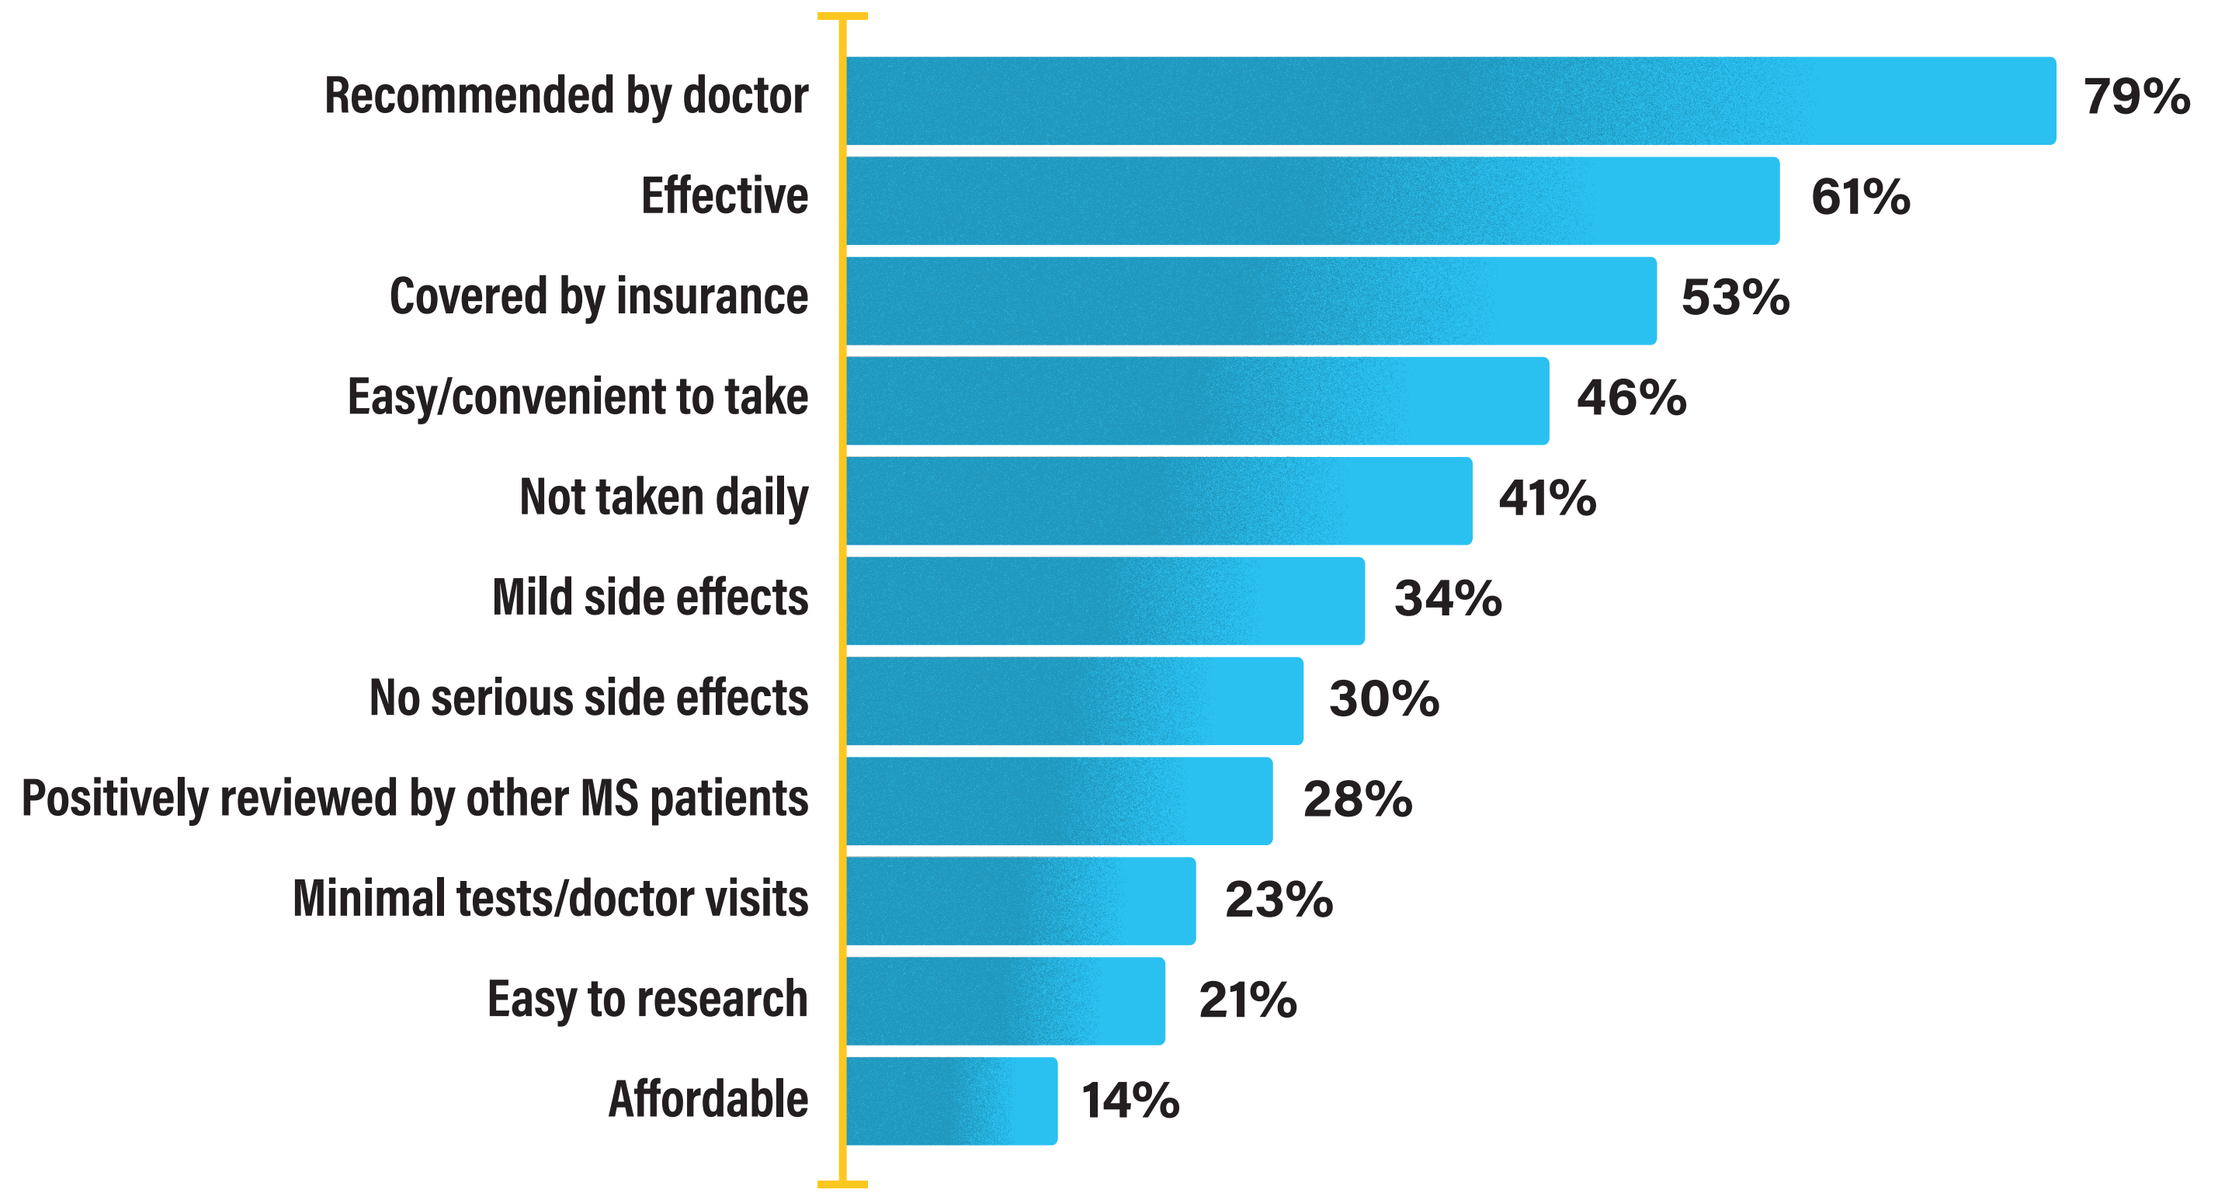
Recommended by doctor 79%
Effective 61%
Covered by insurance 53%
Easy/convenient to take 46%
Not taken daily 41%
Mild side effects 34%
No serious side effects 30%
Positively reviewed by other MS patients 28%
Minimal tests/doctor visits 23%
Easy to research 21%
Affordable 14%

Q. When you made the decision to go on your most recent DMT medication, which of the following factors were most important to you in your decision? Select all that apply? (Asked of those currently on a DMT) (Sample Size = 771)

FIGURE 7. Benefits Received From Current DMT

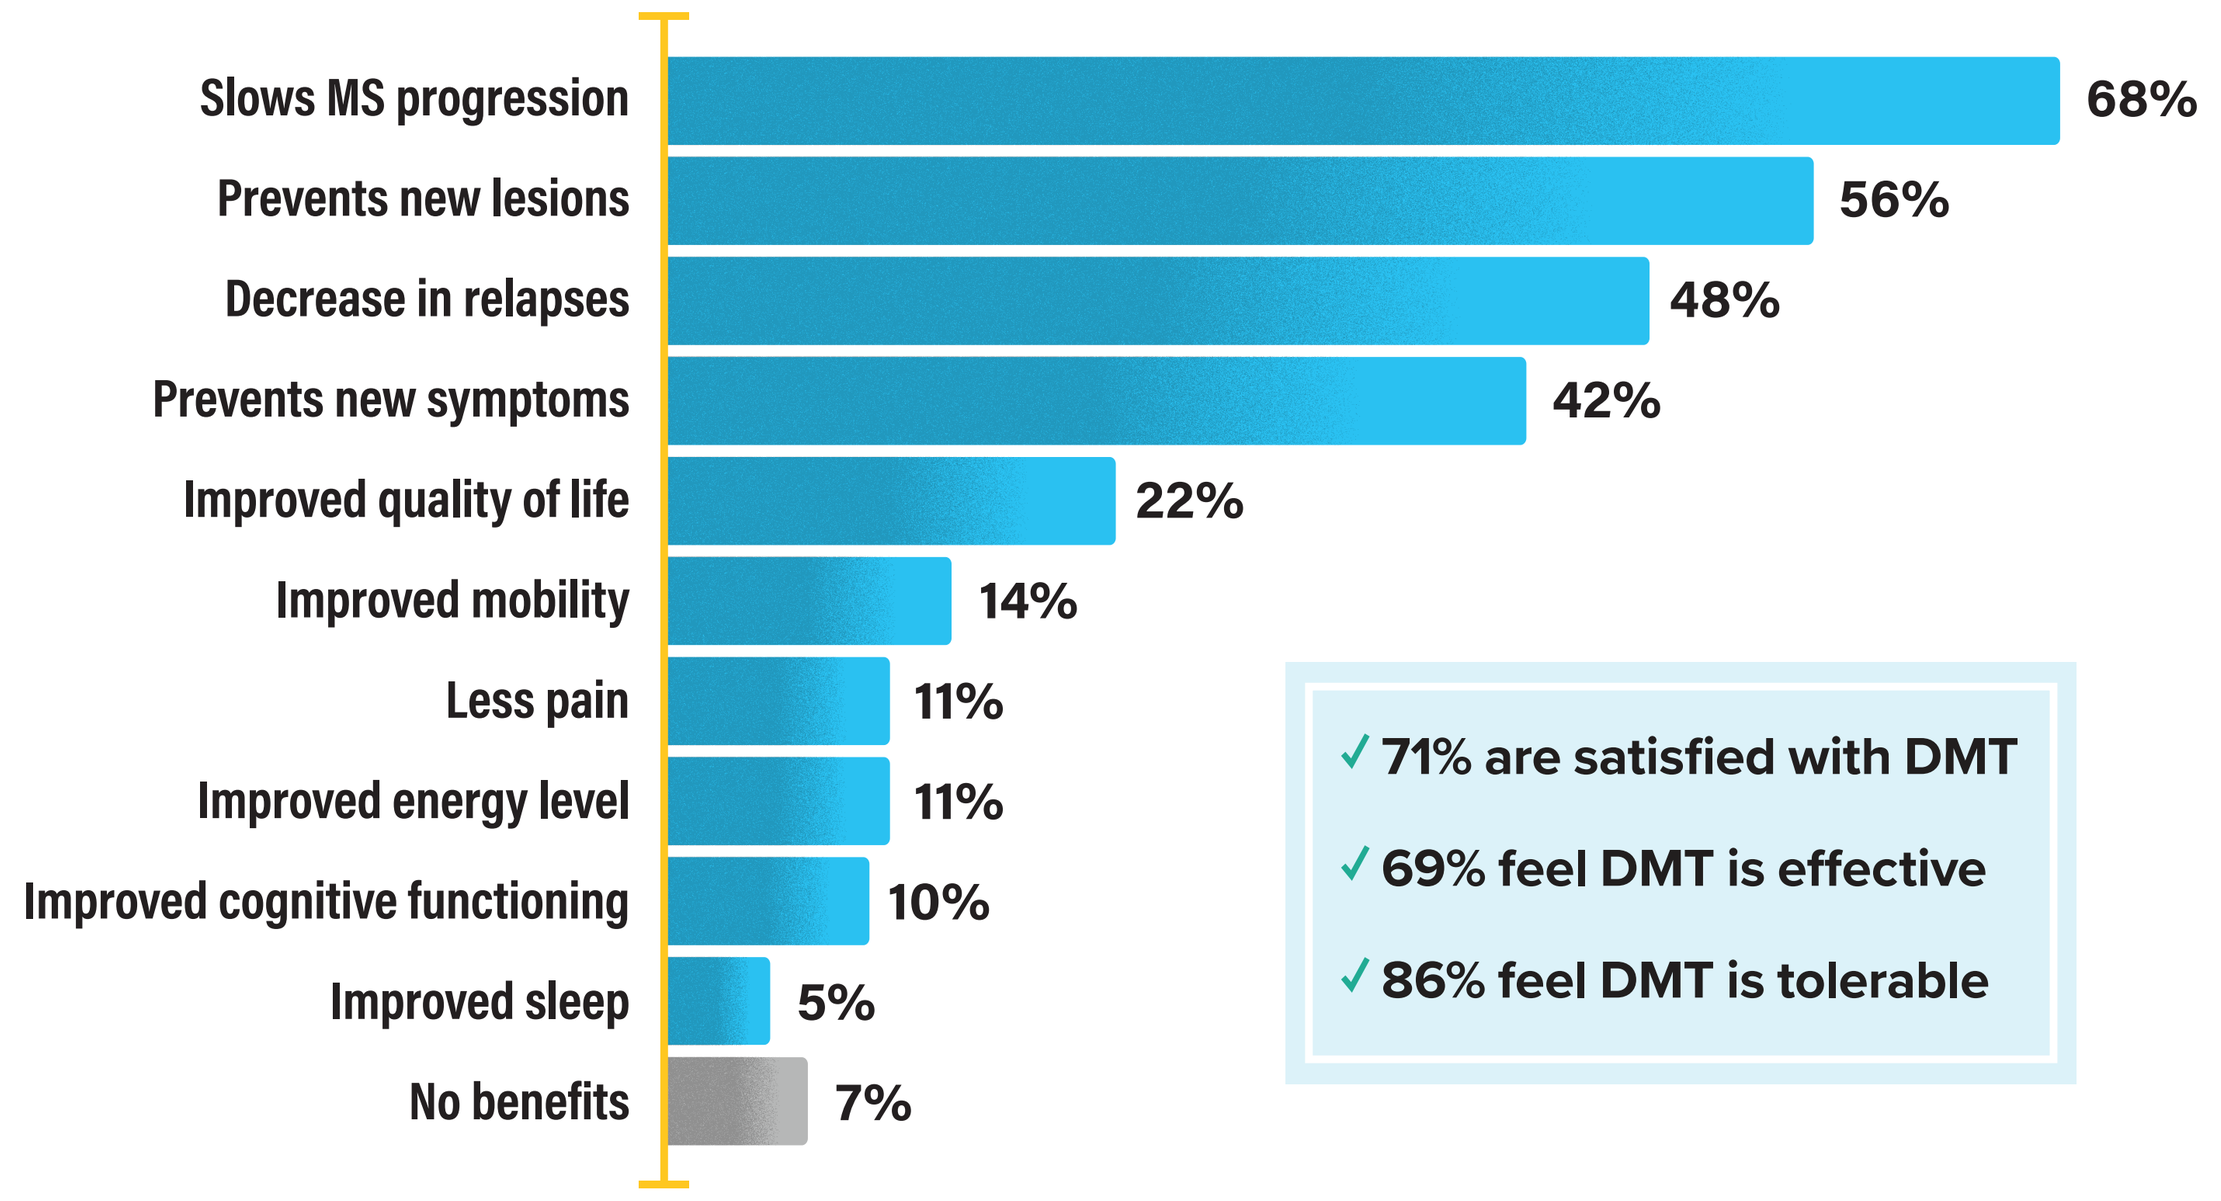
Slows MS progression 68%
Prevents new lesions 56%
Decrease in relapses 48%
Prevents new symptoms 42%
Improved quality of life 22%
Improved mobility 14%
Less pain 11%
Improved energy level 11%
Improved cognitive functioning 10%
Improved sleep 5%
No benefits 7%
✓ 71% are satisfied with DMT
✓ 69% feel DMT is effective
✓ 86% feel DMT is tolerable

Q. What are the main benefits you receive from your current DMT? Select all that apply. (Asked of those currently on a DMT) (Sample Size = 771)

FIGURE 8. Top Three Desired DMT Benefits

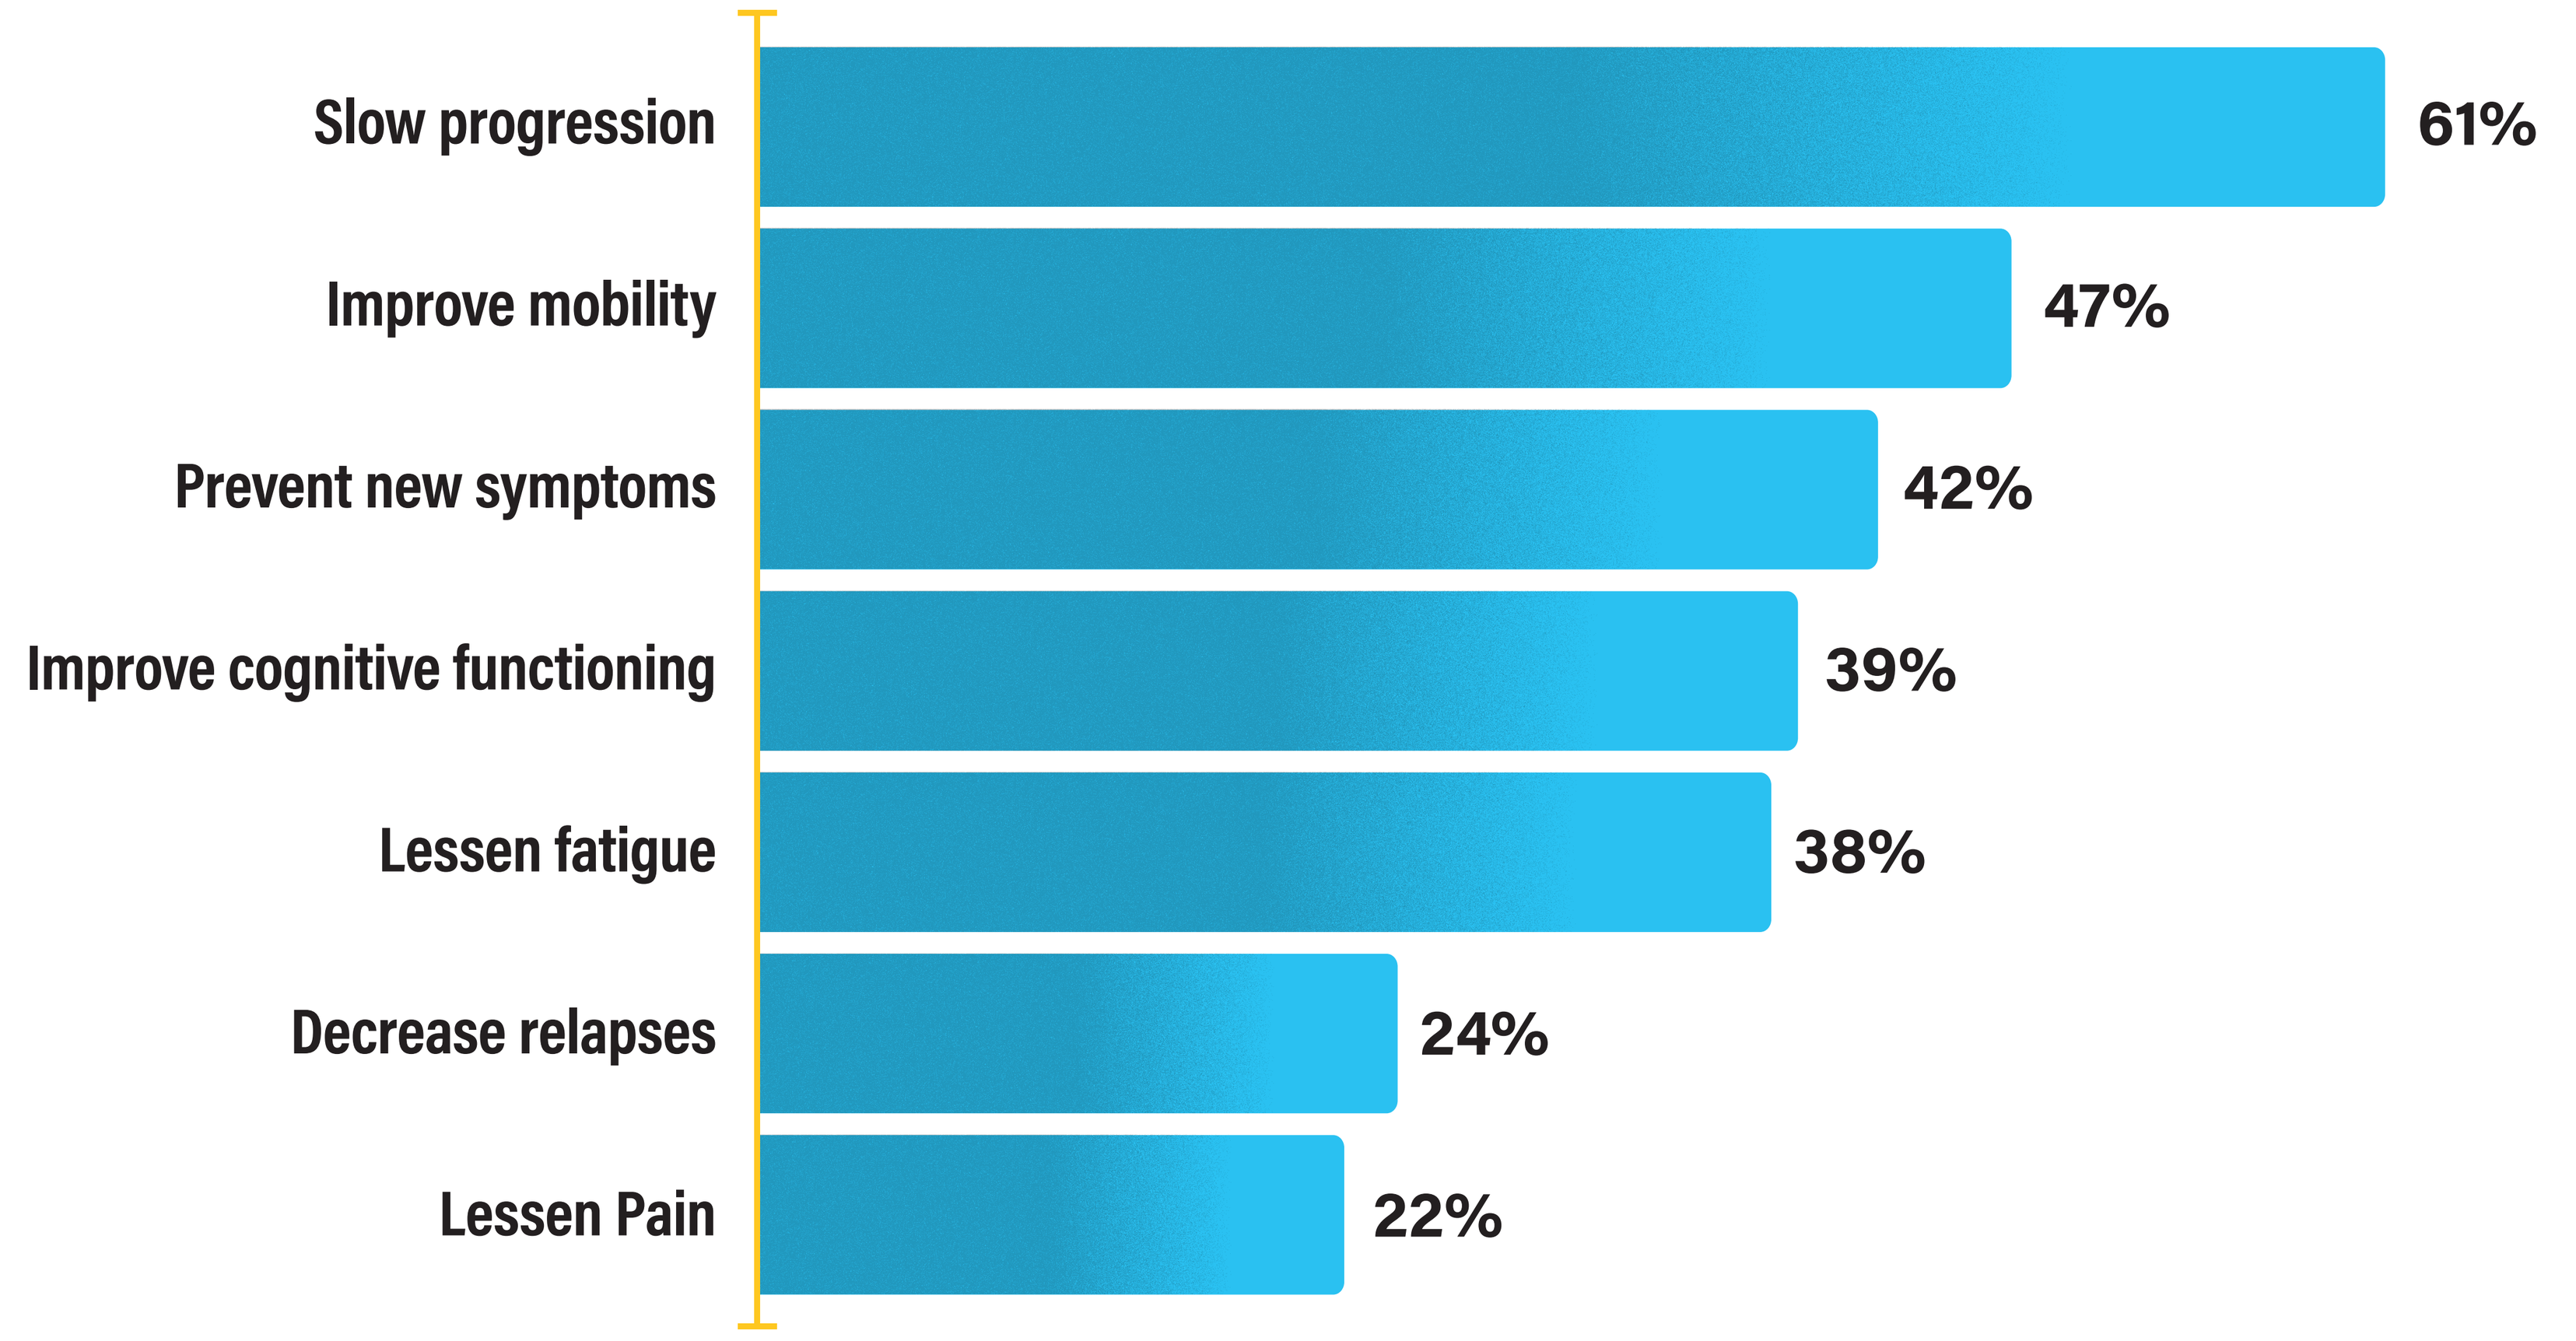
Slow progression 61%
Improve mobility 47%
Prevent new symptoms 42%
Improve cognitive functioning 39%
Lessen fatigue 38%
Decrease relapses 24%
Lessen Pain 22%

Q. When considering a DMT's effectiveness, what is it you most want it to address? Select up to your top 3 choices. (Sample Size = 1294)

FIGURE 9. Reasons Might Try a DMT (DMT Naïve)

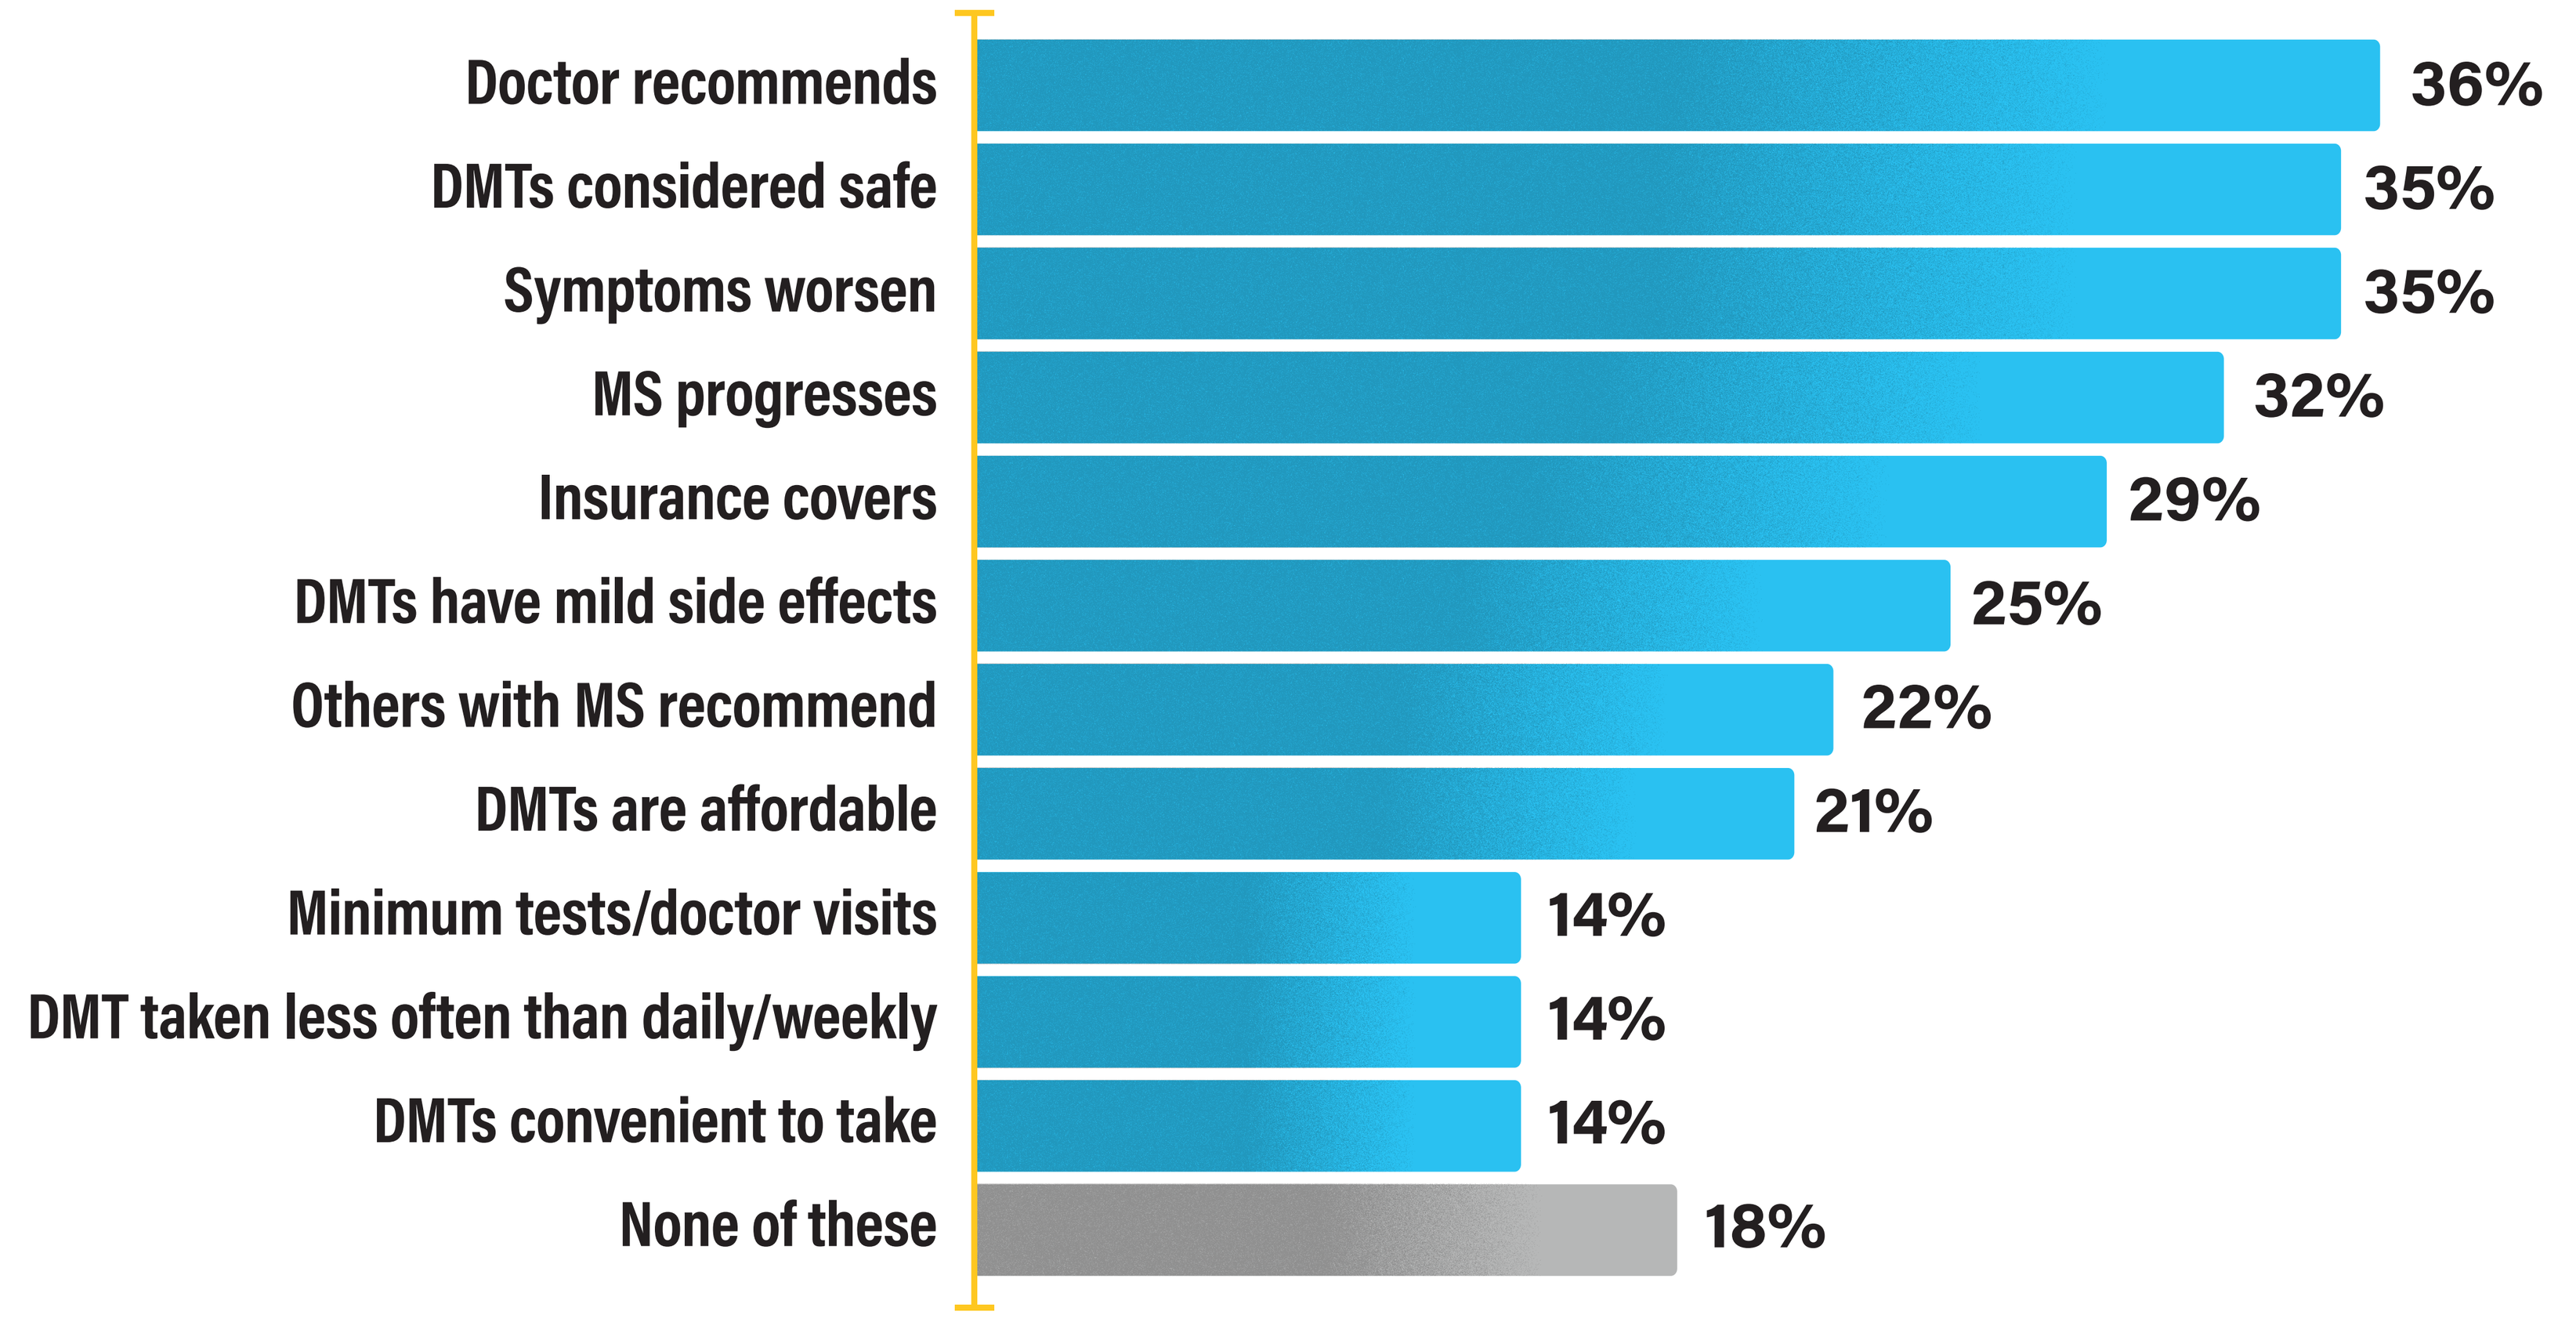
Doctor recommends 36%
DMTs considered safe 35%
Symptoms worsen 35%
MS progresses 32%
Insurance covers 29%
DMTs have mild side effects 25%
Others with MS recommend 22%
DMTs are affordable 21%
Minimum tests/doctor visits 14%
DMT taken less often than daily/weekly 14%
DMTs convenient to take 14%
None of these 18%

Q. Which, if any, of the following might be reasons why you would start taking prescription DMTs in the future? Select all that apply. (Asked of those never on a DMT) (Sample Size = 164)